Efficacy of ND0612 for nocturnal problems and early morning OFF: A blinded rater study of 2 dosing regimens

Alberto Espay[1], Aaron Ellenbogen[2], Sheila Oren[3], Ryan Case[3], Karl Kieburtz[4], C. Warren Olanow[4]

[1]University of Cincinnati, Cincinnati, OH; [2]Michigan Institute for Neurological Disorders, Farmington Hills, MI; [3]NeuroDerm Ltd., Rehovot, Israel; [4]Clintrex, Sarasota, FL

Introduction

- Nocturnal symptoms and early morning OFF (EMO) periods are significant contributors to poor quality of life in patients with Parkinson's disease (PD) experiencing motor fluctuations.[1, 2] Patients suffer a variety of problems including difficulty turning in bed, restless legs and sleep fragmentation, as well as early morning akinesia, dystonia, and tremor.[2]
- ND0612 is a drug-device combination that continuously delivers liquid levodopa/carbidopa (60/7.5 mg/mL) by subcutaneous infusion through a non-surgical mini-pump system to reduce motor complications in PD. Phase 1 & 2 trials have demonstrated that ND0612 maintained steady, therapeutic levodopa plasma concentrations that were associated with reduced OFF time.[3]
- We have previously reported the primary efficacy results from this Phase-2 study (NCT02577523) which showed that continuous delivery of ND0612 reduced total daily OFF time.[4] In this analysis of secondary outcomes, we focus on the benefits of treatment on nocturnal symptoms and EMO periods.

Methods

- This was a 28-day randomized, parallel-group, open label, blinded-rater study.
- Outcomes of interest include Change from baseline to endpoint in:
 - Percent of subjects with full ON at 8 & 9 AM (Key secondary endpoint)
 - Night-time sleep quality, as assessed using PDSS-2
 - Subjective patient assessment of sleep quality (5 point scale)
 - Early morning (8 AM) UPDRS motor scores (*post-hoc* analysis).

Study design

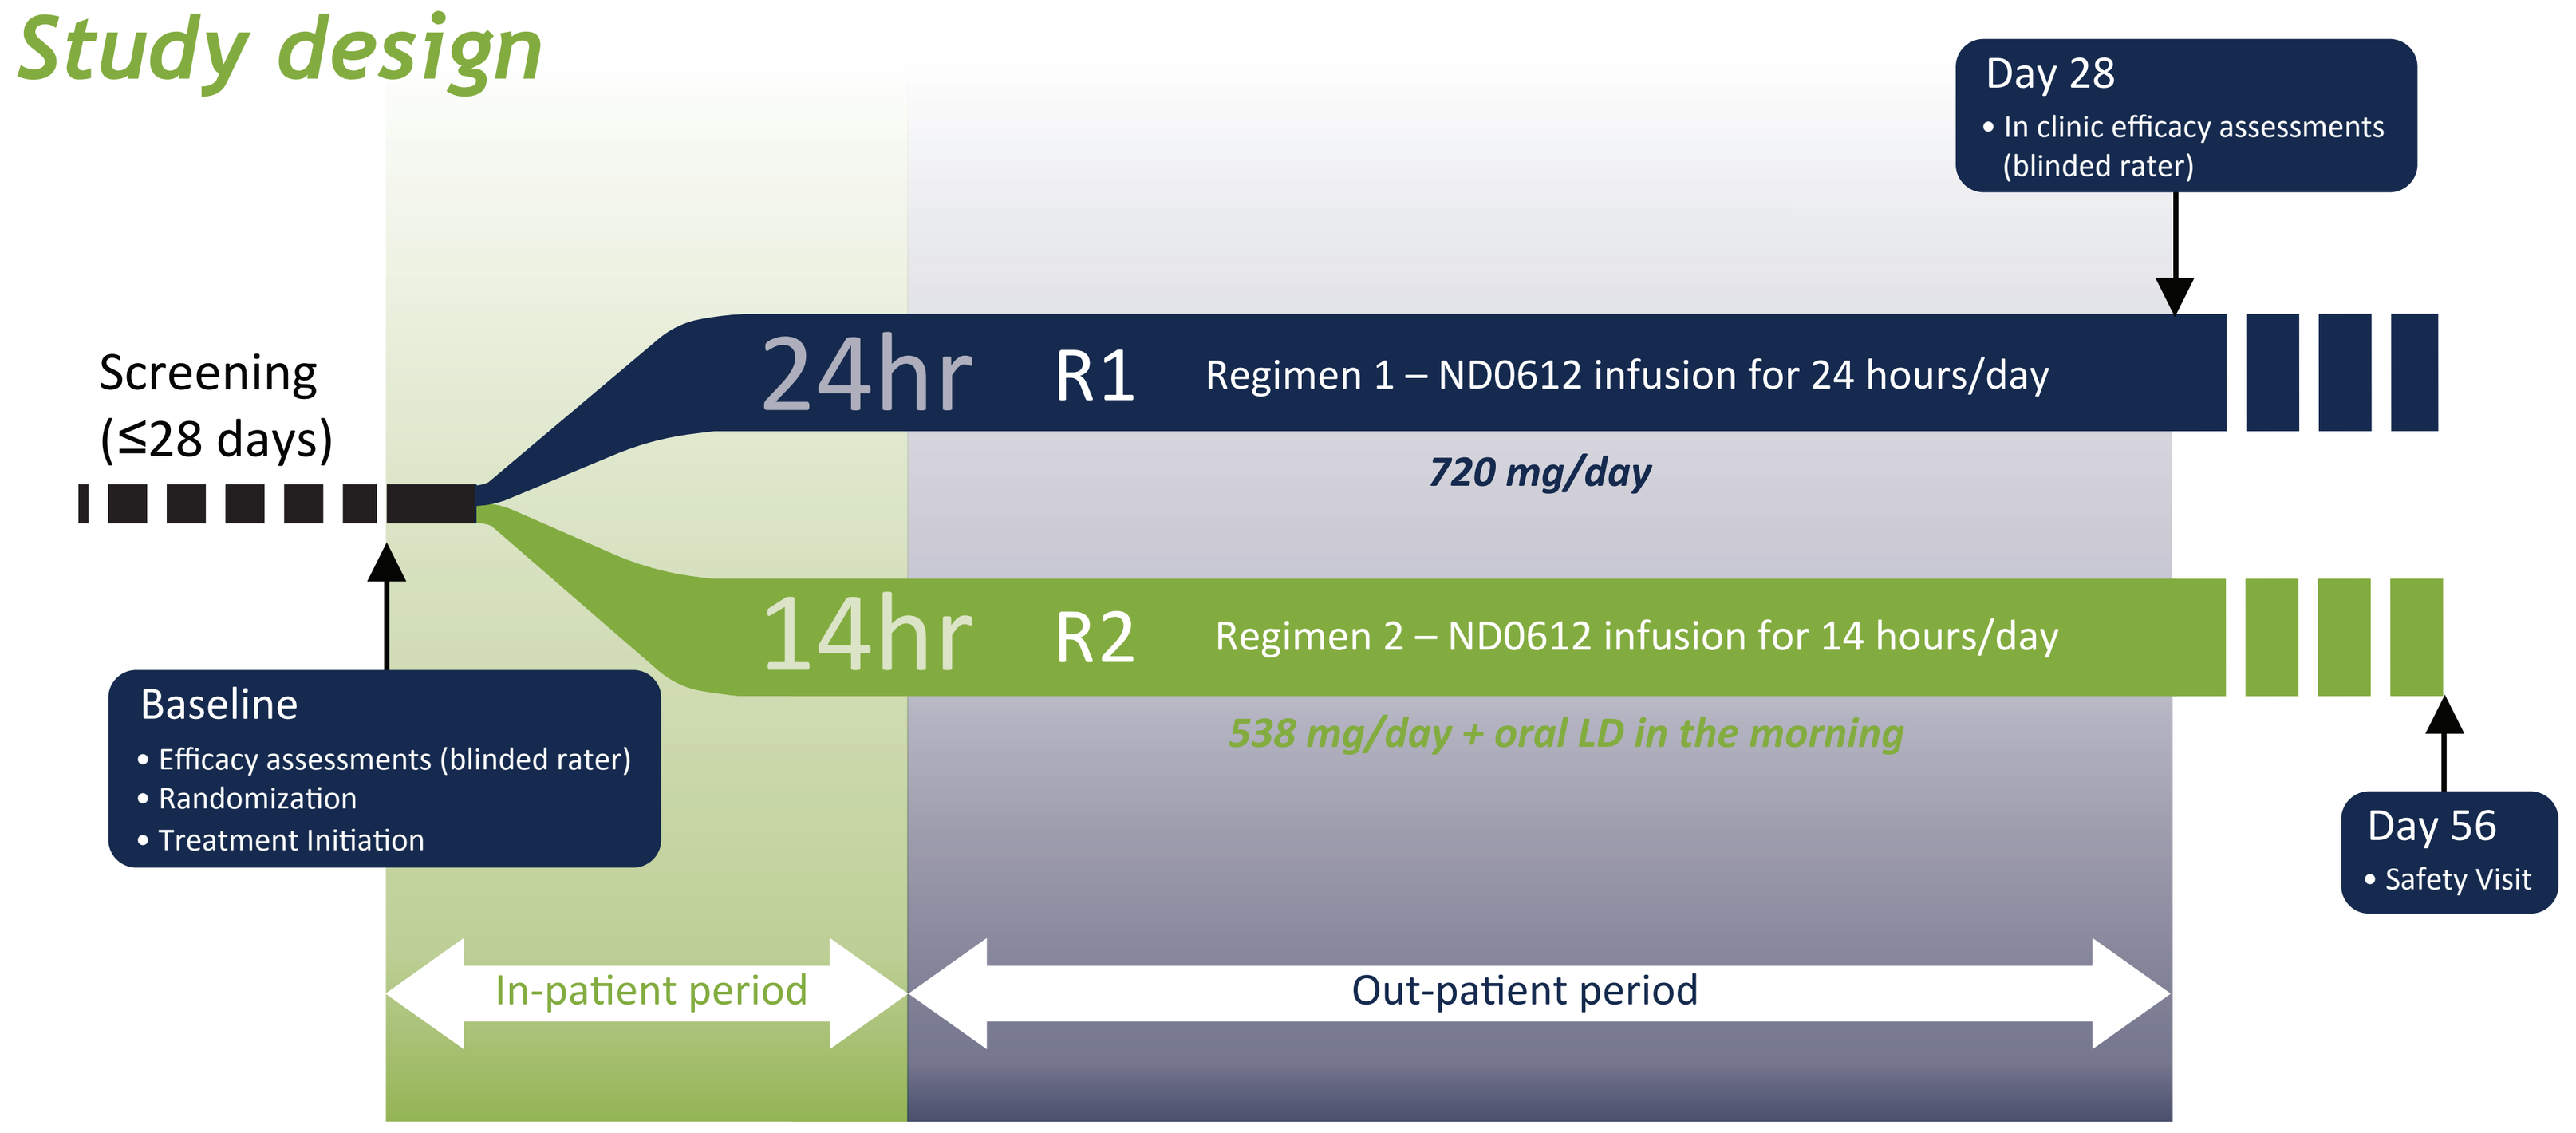
Screening
(≤28 days)
Baseline
• Efficacy assessments (blinded rater)
• Randomization
• Treatment Initiation
24hr R1 Regimen 1 – ND0612 infusion for 24 hours/day
720 mg/day
14hr R2 Regimen 2 – ND0612 infusion for 14 hours/day
538 mg/day + oral LD in the morning
Day 28
• In clinic efficacy assessments (blinded rater)
Day 56
• Safety Visit
In-patient period
Out-patient period

Male and female (30 – 80 years) patients with a diagnosis of PD and a Hoehn & Yahr stage ≤3 (during ON) were eligible for the study. Patients had to be taking ≥4 levodopa doses per day (≥3 doses/day of IPX-066), have ≥2.5 hours of OFF time per day including predictable and well defined EMO periods, and a 'good' early morning response to levodopa. A total of 38 subjects were randomized (1:1, n=19 in both groups) to 2 dosing regimens of ND0612: R1 (24h infusion with ND0612, total LD/CD dose of 720/90 mg) or R2 (14h 'waking-day' infusion with ND0612, total LD/CD dose of 538/68mg + morning oral LD/CD 150/15 mg). Supplemental oral LD/CD was used as needed.

Results

The proportion of subjects with full ON increased at 8AM and 9AM with 24 hour dosing (R1)

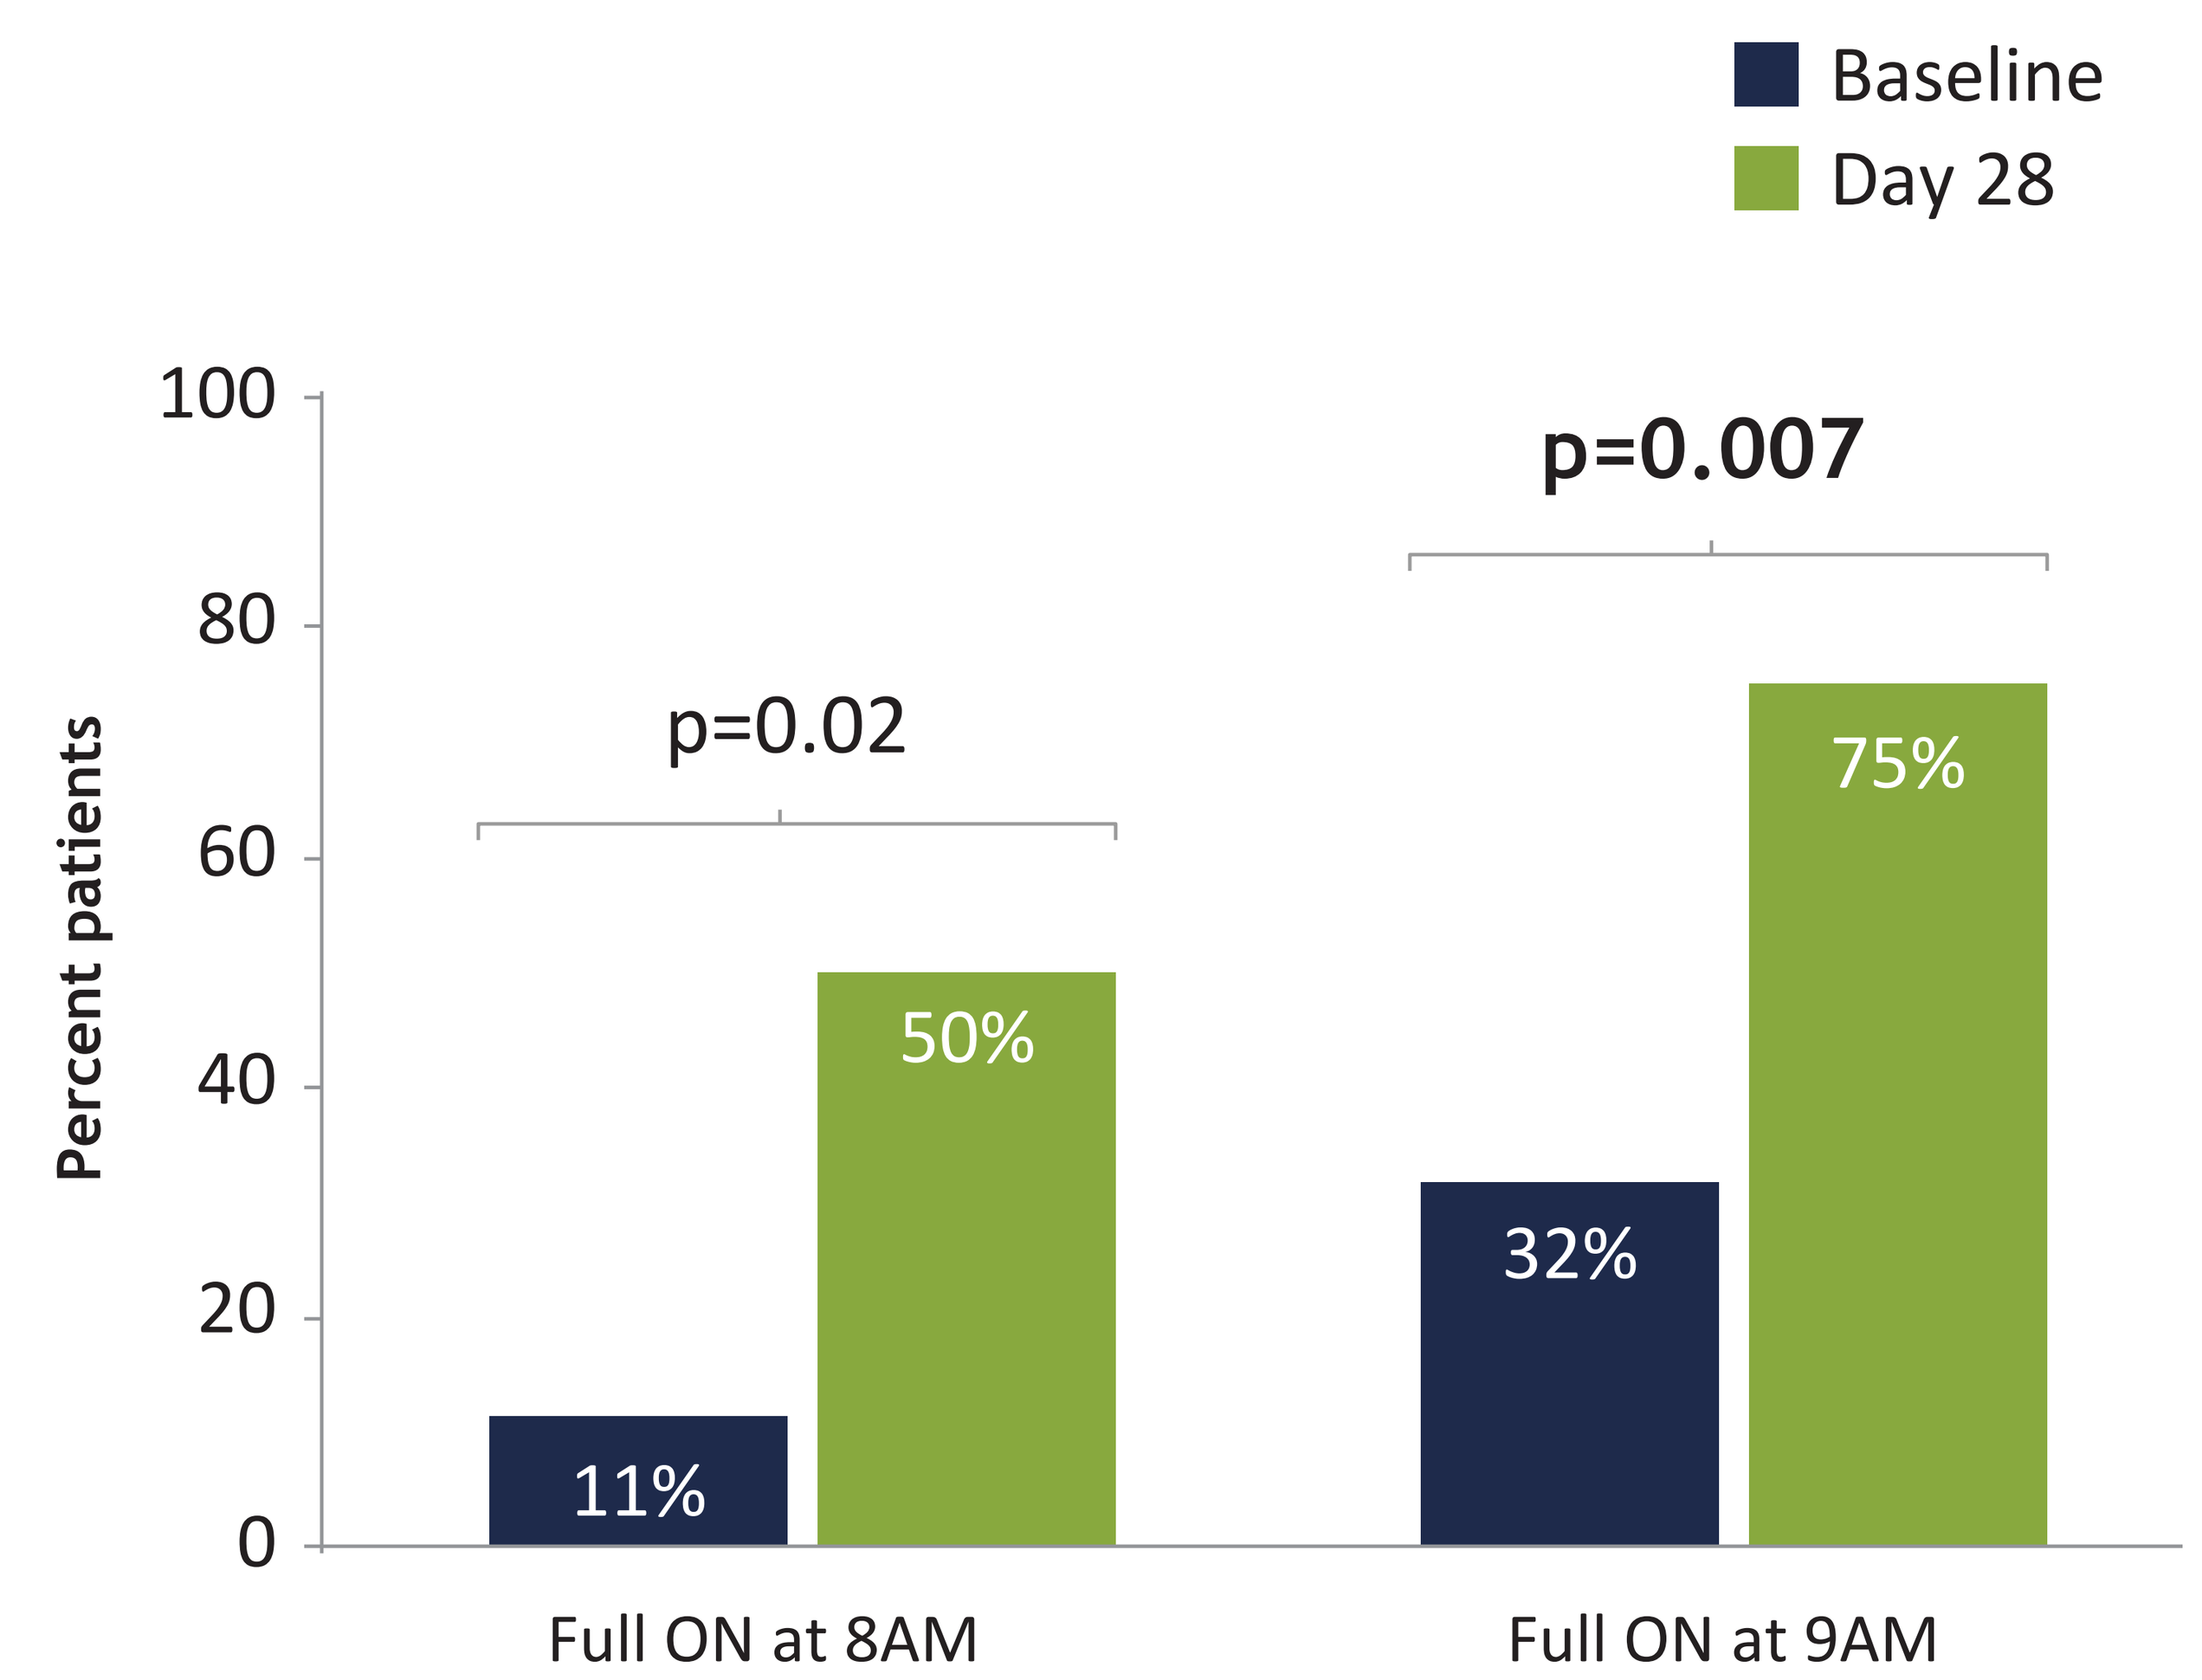
Baseline
Day 28
Percent patients
100
80
60
40
20
0
p=0.02
p=0.007
11%
50%
32%
75%
Full ON at 8AM
Full ON at 9AM

In R2, dosing began later in the morning and thus no improvement in the proportion of patients with full ON was seen at 8 & 9 AM

Early morning (8 AM) UPDRS motor scores significantly improved in both groups

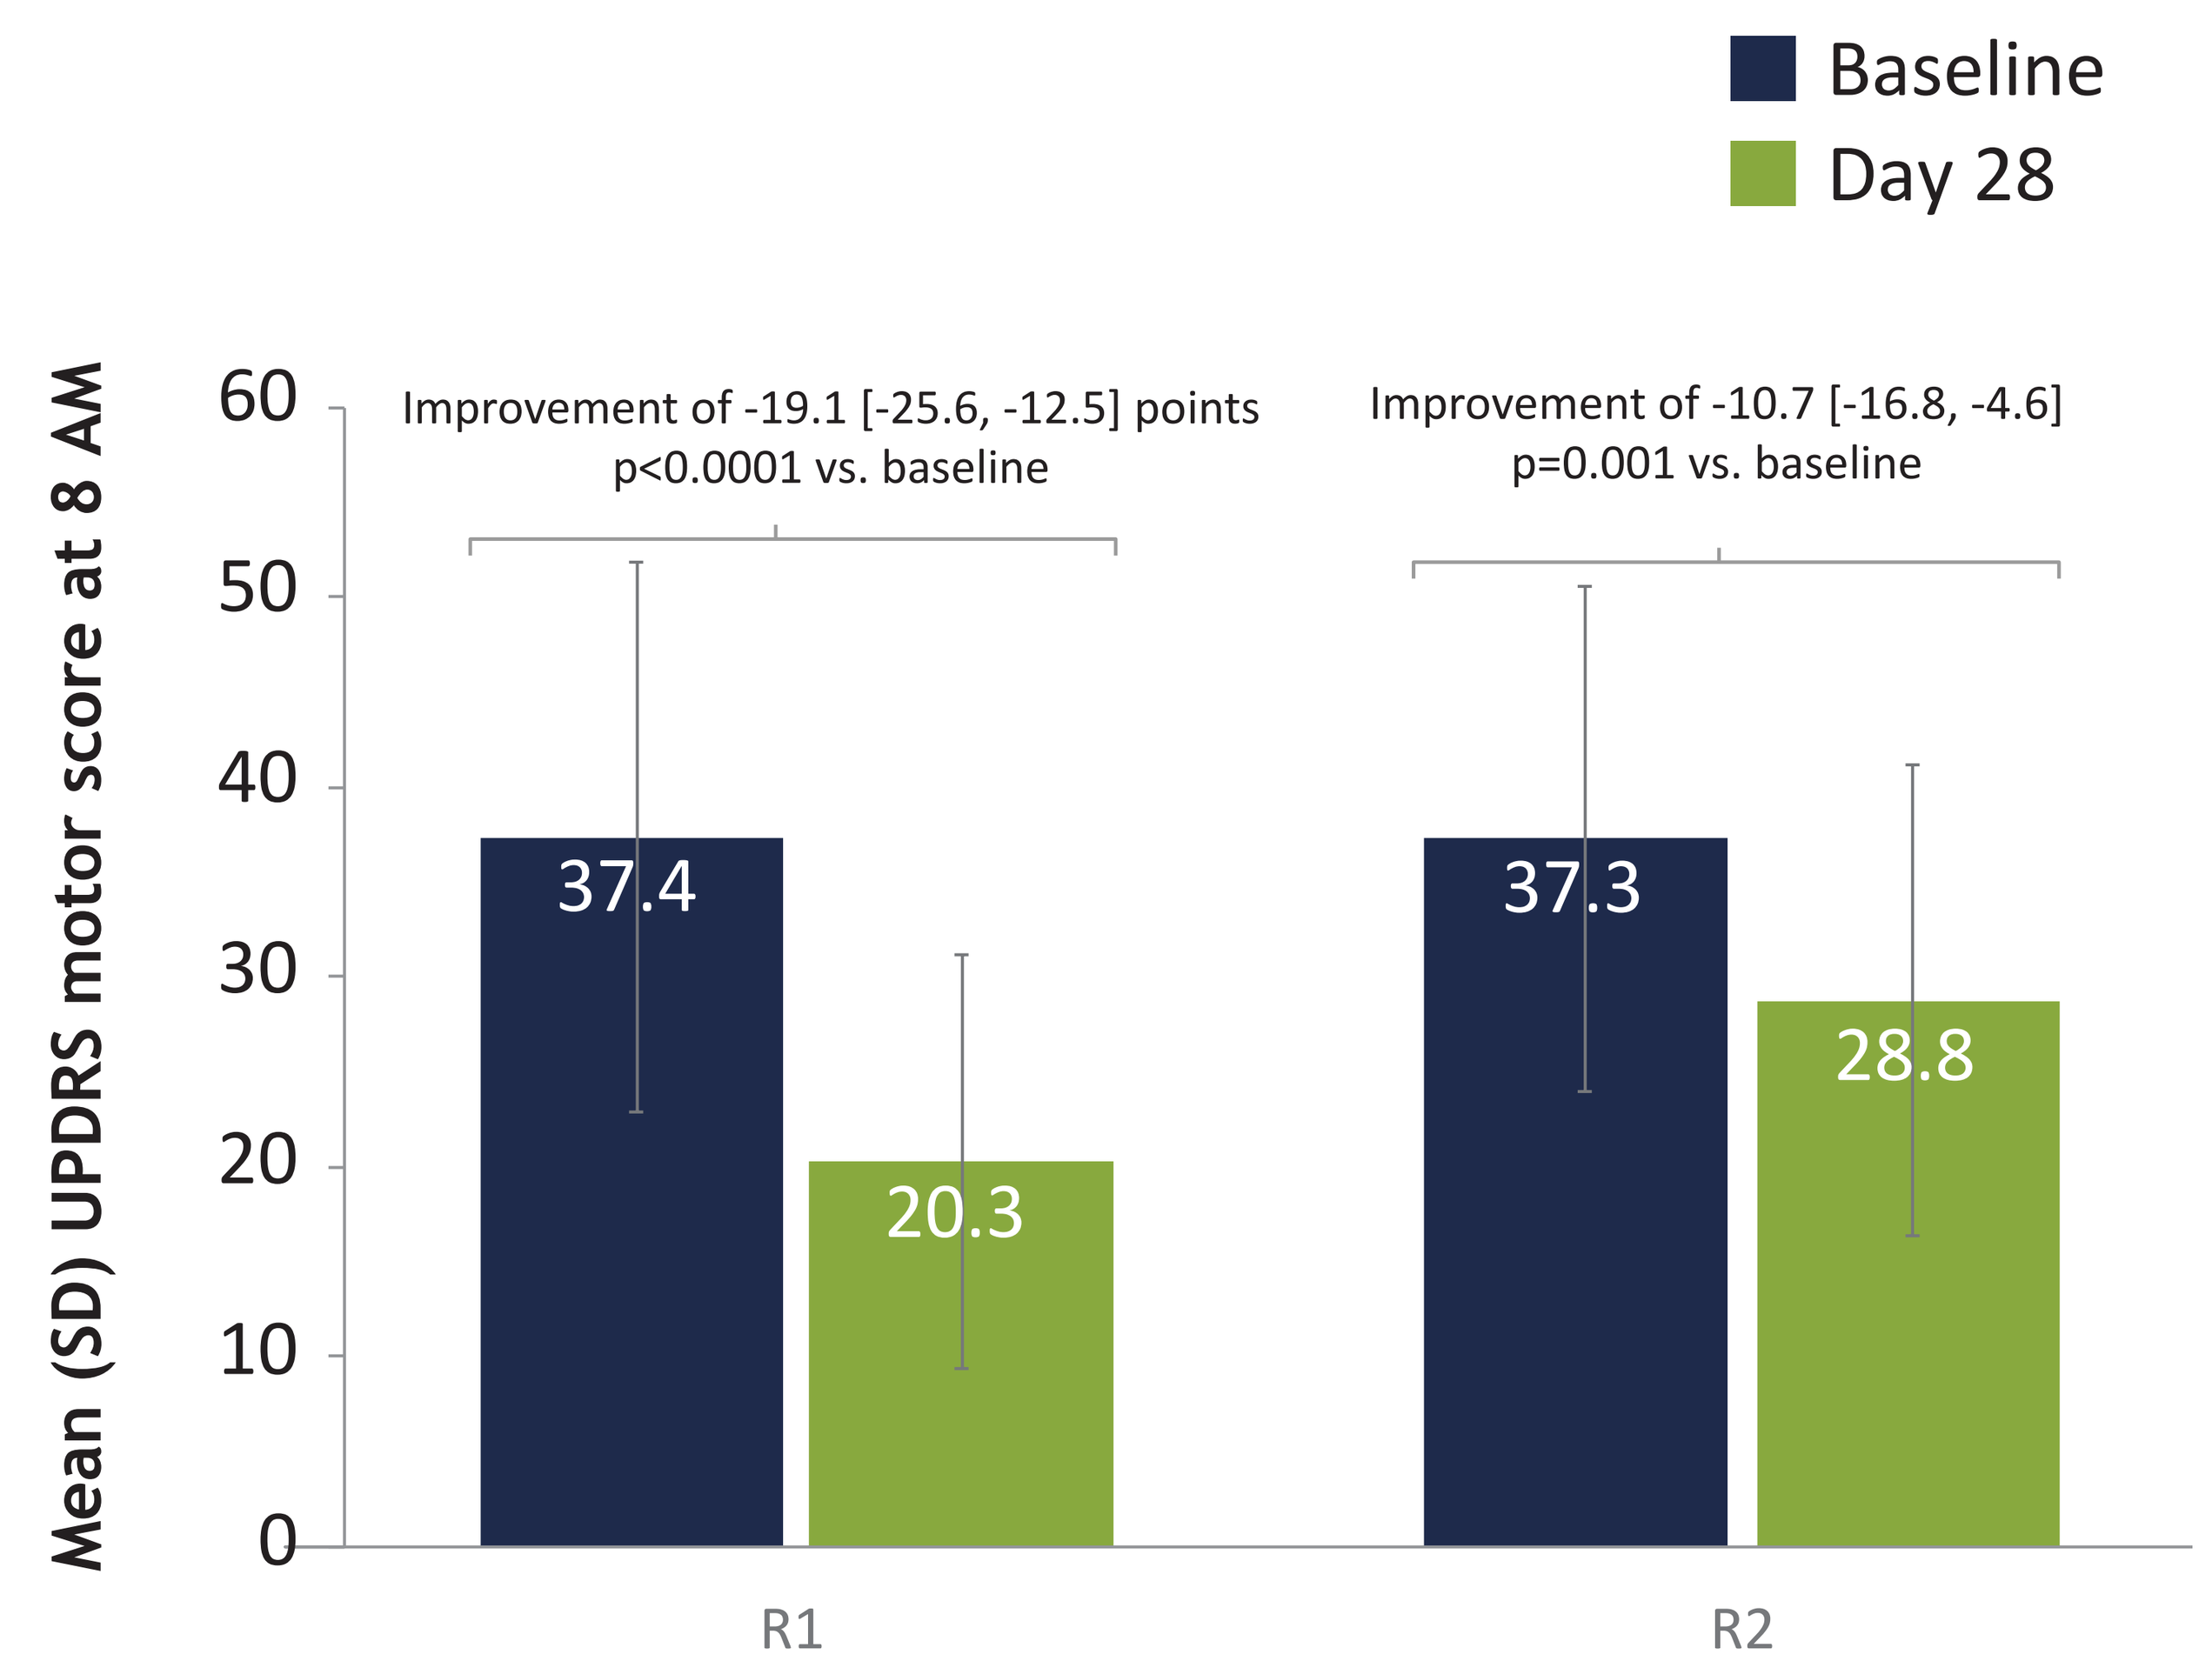
Baseline
Day 28
Mean (SD) UPDRS motor score at 8 AM
60
50
40
30
20
10
0
Improvement of -19.1 [-25.6, -12.5] points
p<0.0001 vs. baseline
Improvement of -10.7 [-16.8, -4.6]
p=0.001 vs. baseline
37.4
20.3
37.3
28.8
R1
R2

Change from baseline data are mean [95% CI].

PDSS-2 scores significantly improved from baseline with 24-hour dosing (R1)

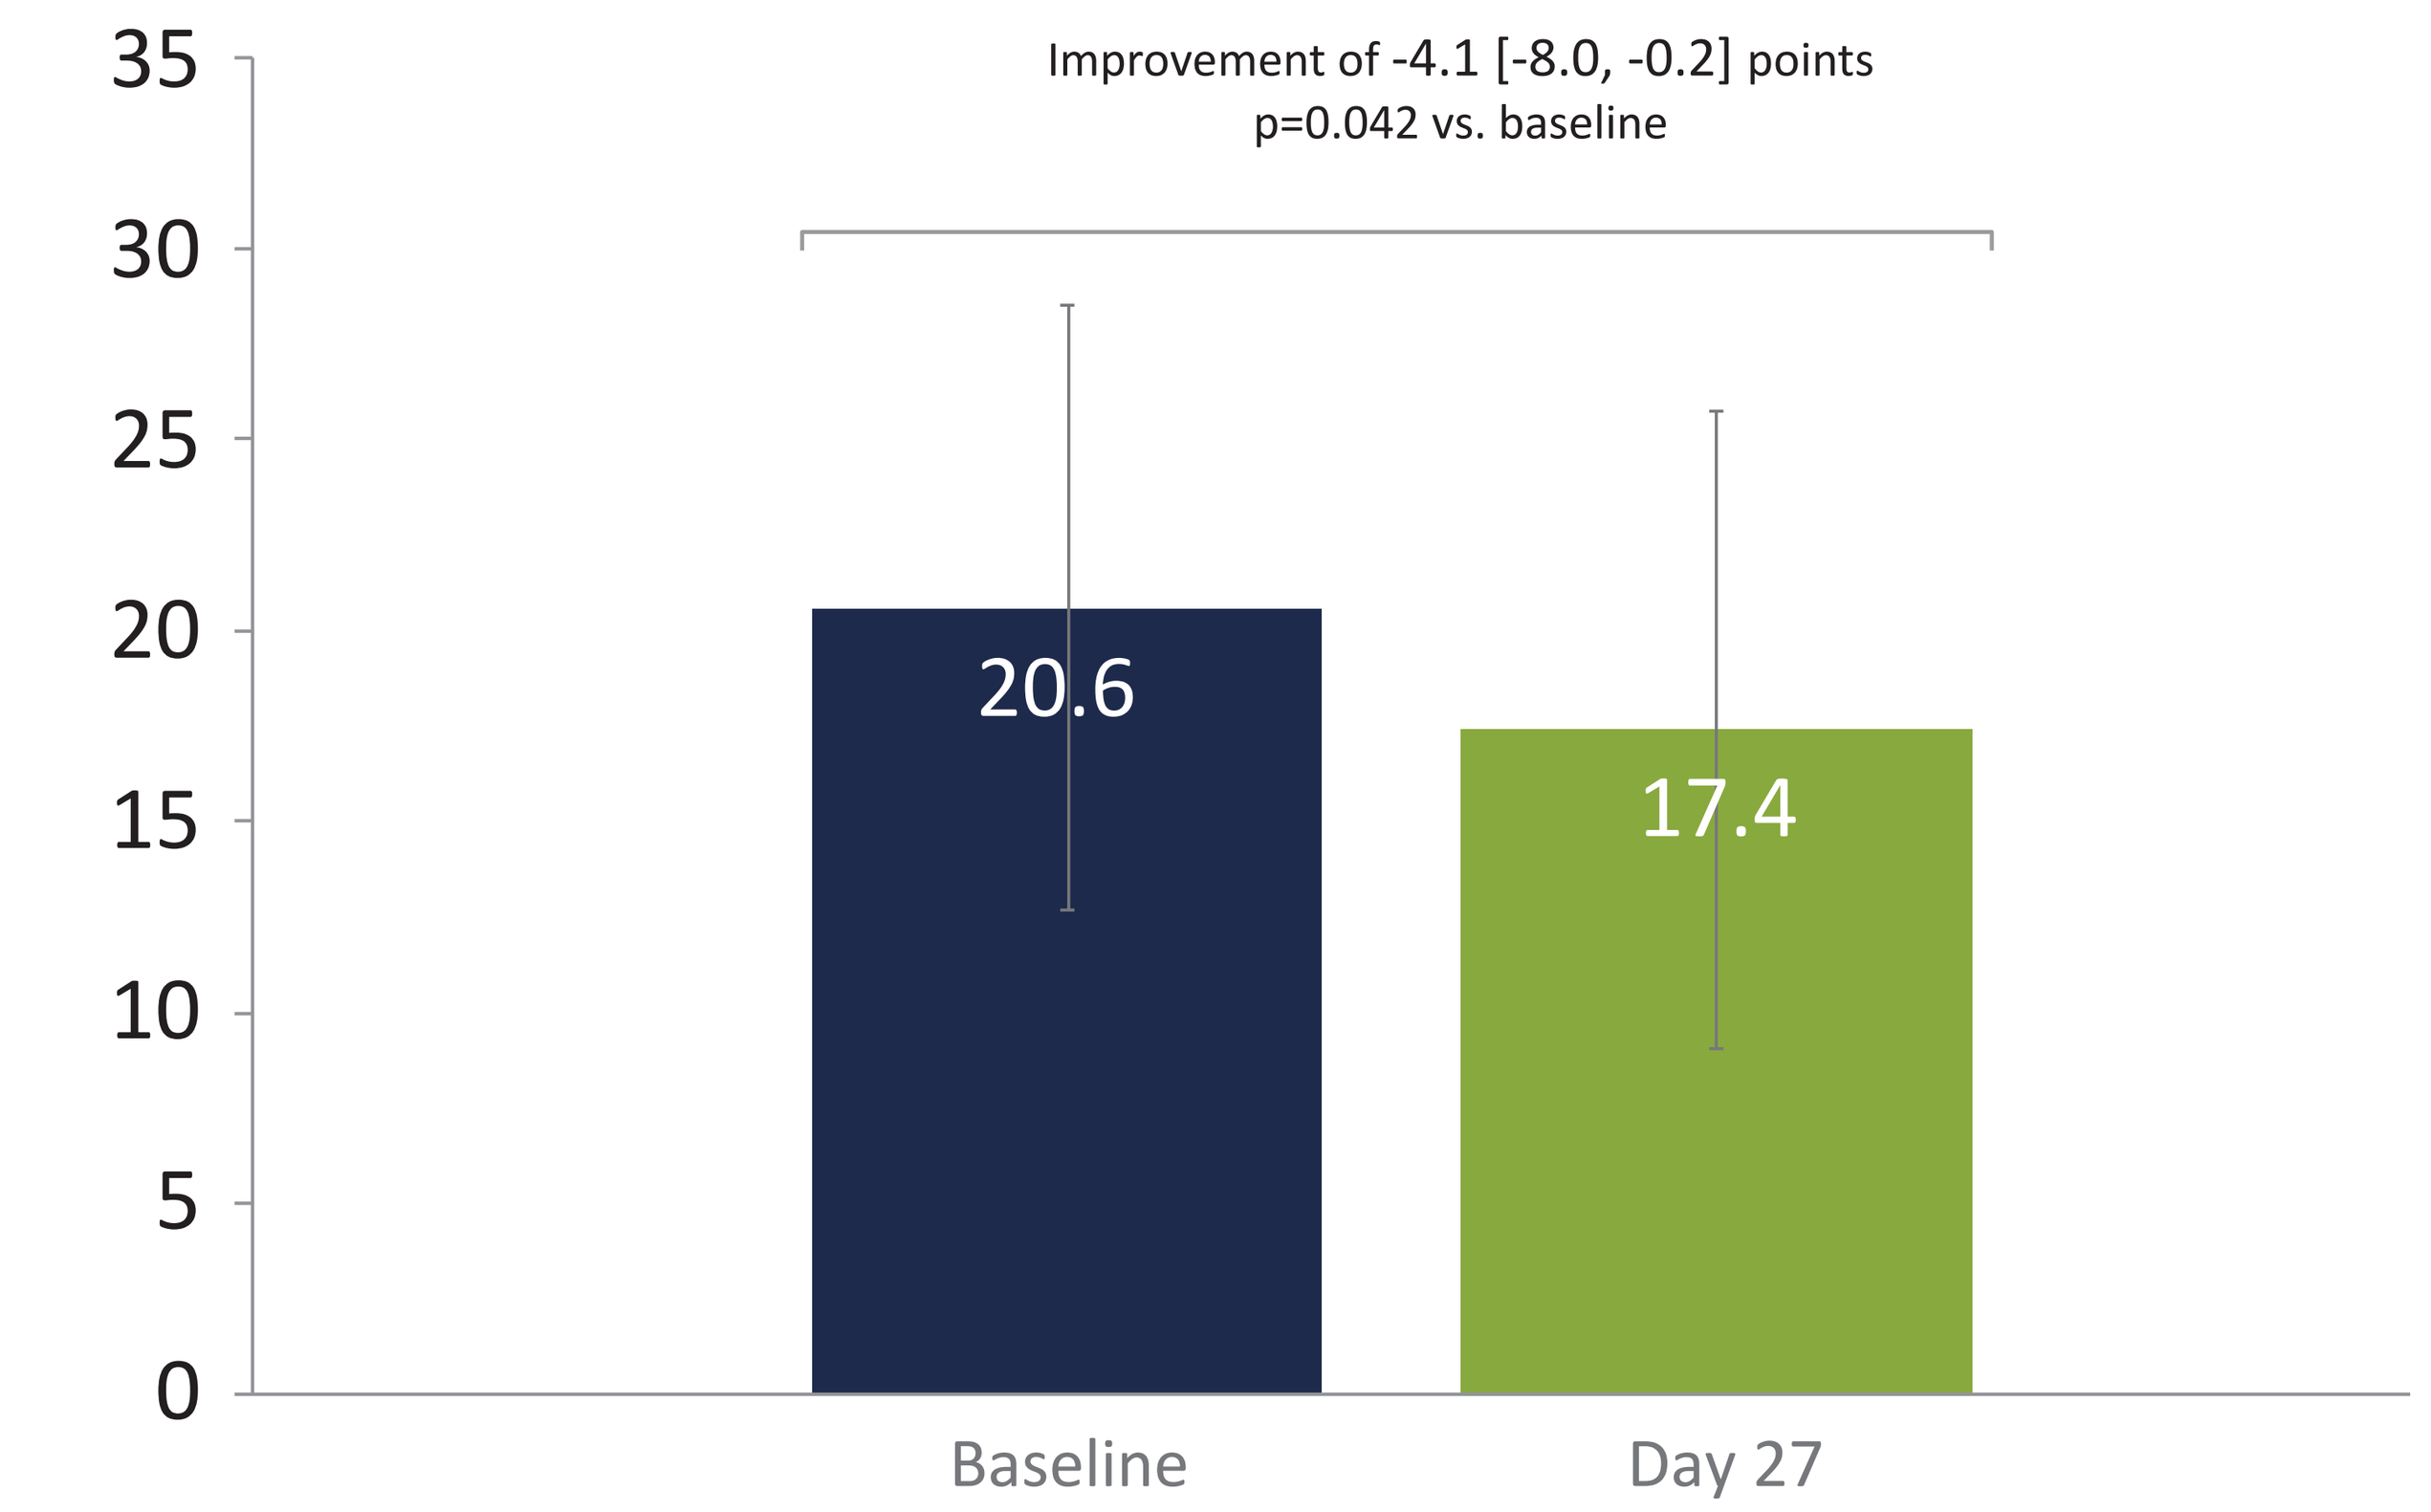
35
30
25
20
15
10
5
0
Improvement of -4.1 [-8.0, -0.2] points
p=0.042 vs. baseline
20.6
17.4
Baseline
Day 27

Change from baseline data are mean [95% CI]. Waking day infusion (R2) had no significant effect on nocturnal sleep quality.

Treatment with R1 improved patient ratings of sleep quality

Patients self-rated the statement *"I slept well last night"*

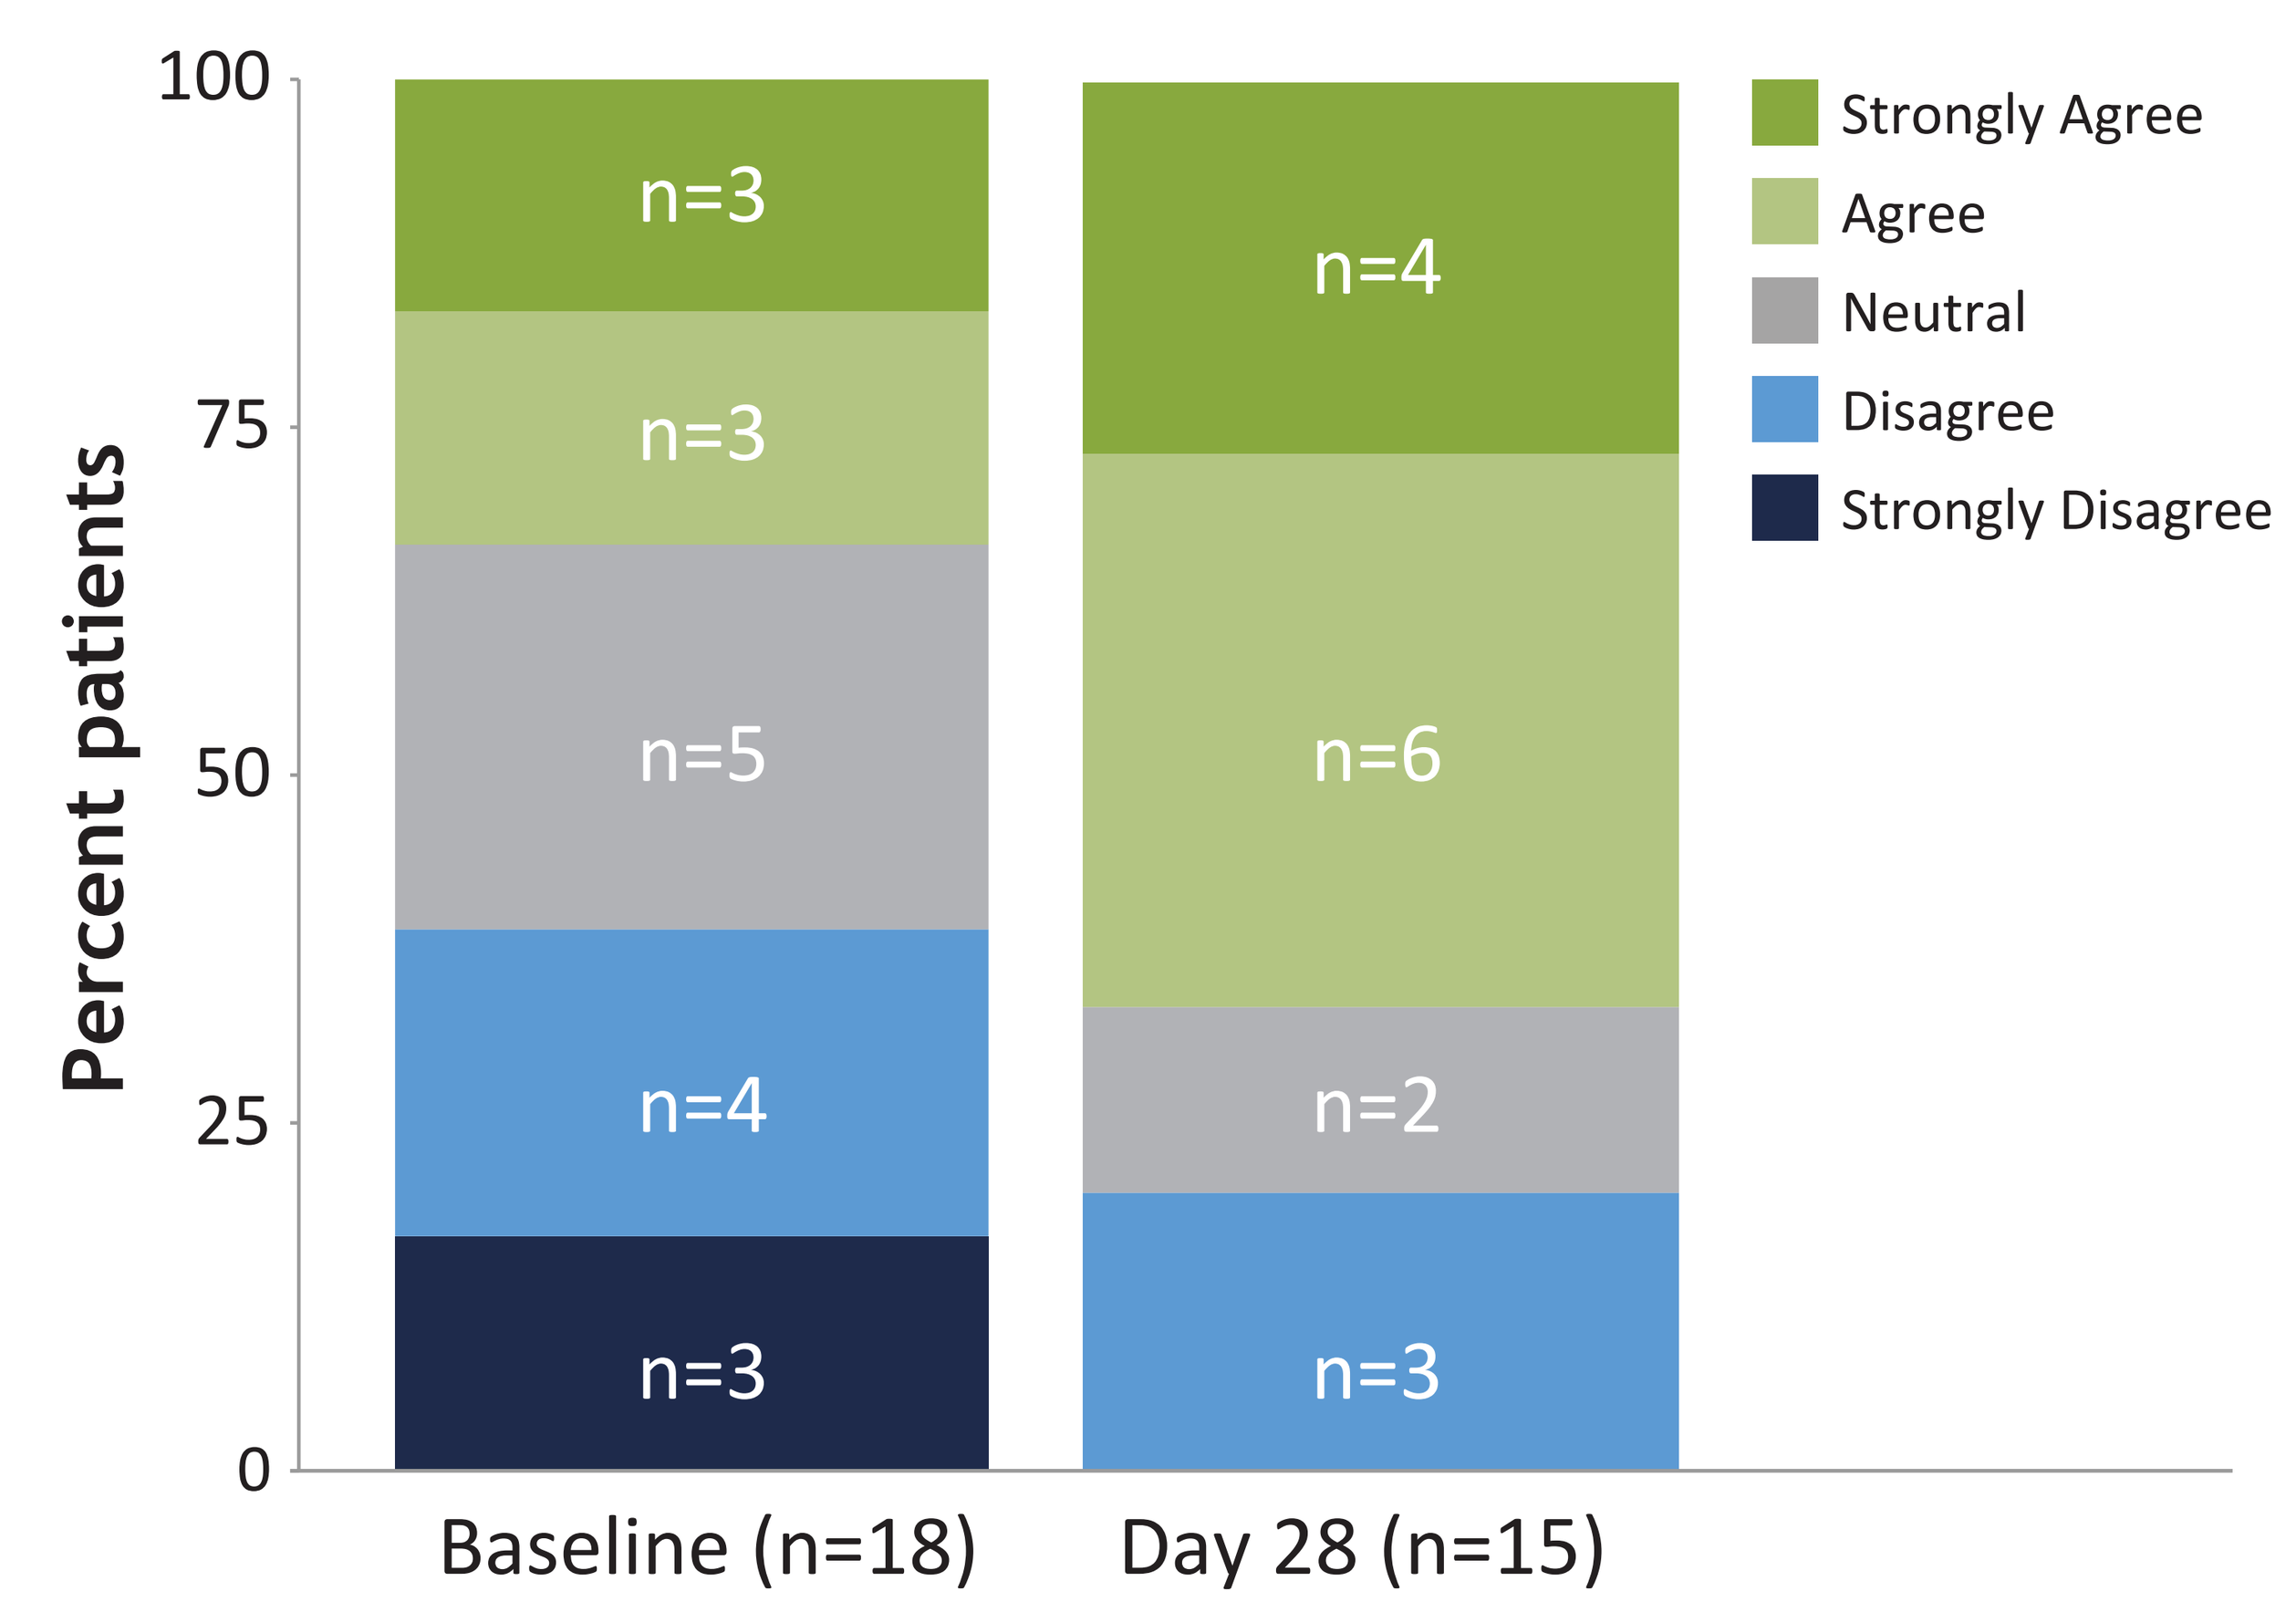
Percent patients
100
75
50
25
0
n=3
n=3
n=5
n=4
n=3
n=4
n=6
n=2
n=3
Baseline (n=18)
Day 28 (n=15)
Strongly Agree
Agree
Neutral
Disagree
Strongly Disagree

Width of bars are proportional to number of patients with data available. Overall, 50% of R1 patients reported an improvement in sleep with R1. Patients in R2 did not have overnight infusion, and 25% of R2 patients reported an improvement in sleep.

Conclusions

- 24 hour, 'round-the-clock' levodopa infusion with ND0612 significantly increased morning ON-time with a relevant improvement in motor status.
- Sleep quality also improved, indicating that patients tolerated the pump overnight.
- In this study, R2 (waking day dosing) was not optimized for nighttime and early morning use because it provided a relatively low daily levodopa dose and daily treatment initiation was delayed until a nurse started the pump. A longer daytime regimen of 16 hours (LD/CD 720/90 mg) starting immediately upon waking is under evaluation.
- ND0612 may provide a novel non-surgical option for continuous 24-hour levodopa delivery in patients with PD experiencing motor fluctuations, in particular nocturnal symptoms and early morning OFF.

References

1. Chapuis S, Ouchchane L, Metz O, Gerbaud L, Durif F. Impact of the motor complications of Parkinson's disease on the quality of life. Mov Disord 2005;20(2):224-230.
2. Chaudhuri KR. Nocturnal symptom complex in PD and its management. Neurology 2003;61(6 Suppl 3):S17-23.
3. Giladi N, Caraco Y, Gurevich T, et al. ND0612 (levodopa/carbidopa for subcutaneous infusion) achieves stable levodopa plasma levels when administered in low and high doses in patients with PD [Abstract]. Mov Disord. 2017; 32 (suppl 2).
4. Poewe W, Stocchi F, Simuni T, et al. A multicenter, parallel-group, rater-blinded, randomized clinical study investigating the efficacy, safety and tolerability of 2 dosing regimens of ND0612. [Abstract]. Mov Disord 2018;33:S92-93.

Disclosures

Alberto Espay and Aaron Ellenbogen were investigators in the 006 study, and they or their institutions have received payment for participation. Clintrex LLC (K. Kieburtz and C. W. Olanow) provided consultancy for this study. S. Oren and R. Case are employed by NeuroDerm. No author has received financial remuneration for the preparation of this report. Assistance for this poster was provided by A. White and A. Patel (funded by NeuroDerm).

NeuroDerm