1393

Identification of the Carbidopa Concentration in Subcutaneously Administered ND0612 (Levodopa/Carbidopa) that Provides Optimal Levodopa Bioavailability

Liat Adar, Tamar Rachmilewitz Minei, Sheila Oren. NeuroDerm Ltd., Rehovot, Israel

Introduction

It is standard practice to co-administer levodopa with the dopa-decarboxylase (DDC) inhibitor carbidopa to inhibit its peripheral metabolism and maximize the amount of levodopa that reaches the brain.[1] Most oral carbidopa/levodopa combinations are currently formulated at a dose ratio of 1:4.[2] However, preclinical studies have shown that continuous subcutaneous delivery of solubilized carbidopa, not only improves the bioavailability of carbidopa, but also improves the pharmacokinetic profile of oral levodopa by extending its elimination half-life and elevating trough plasma concentrations.[3]

The aim of these two open-label studies was to establish the concentration of subcutaneous carbidopa that provides optimal bioavailability of a concomitant fixed concentration of a novel proprietary liquid formulation of levodopa (ND0612) when given continuously via subcutaneous infusion.

Methods

- Two open-label, dose-finding studies were performed to assess pharmacokinetics of ND0612 formulations with fixed levodopa concentration (60 mg/mL) and varying carbidopa concentrations.
- Both studies assessed the impact of carbidopa dosing on the bioavailability of levodopa derived from ND0612L (low dose) and ND0612H (high dose).

Study 1: ND0612-004

- This study characterized the impact of two doses of carbidopa (7.5 mg/mL and 14 mg/mL) on levodopa bioavailability.
- The study was conducted in clinic, and included adult patients (12 male/4 female; mean ±SD age: 63.1 ± 7.2 years) with idiopathic PD (mean ±SD years since diagnosis: 9.3 ±3.9 years) experiencing well-defined morning OFF periods (≥80% of days) and a good levodopa response.
- Patients were randomized to either ND0612L (total levodopa dose of 115mg over 8 hours) or ND0612H (total levodopa dose of 307 mg over 8 hours). The concentration of levodopa was 60 mg/mL in all treatment regimens throughout the study (Figure 2).

Study 1 design

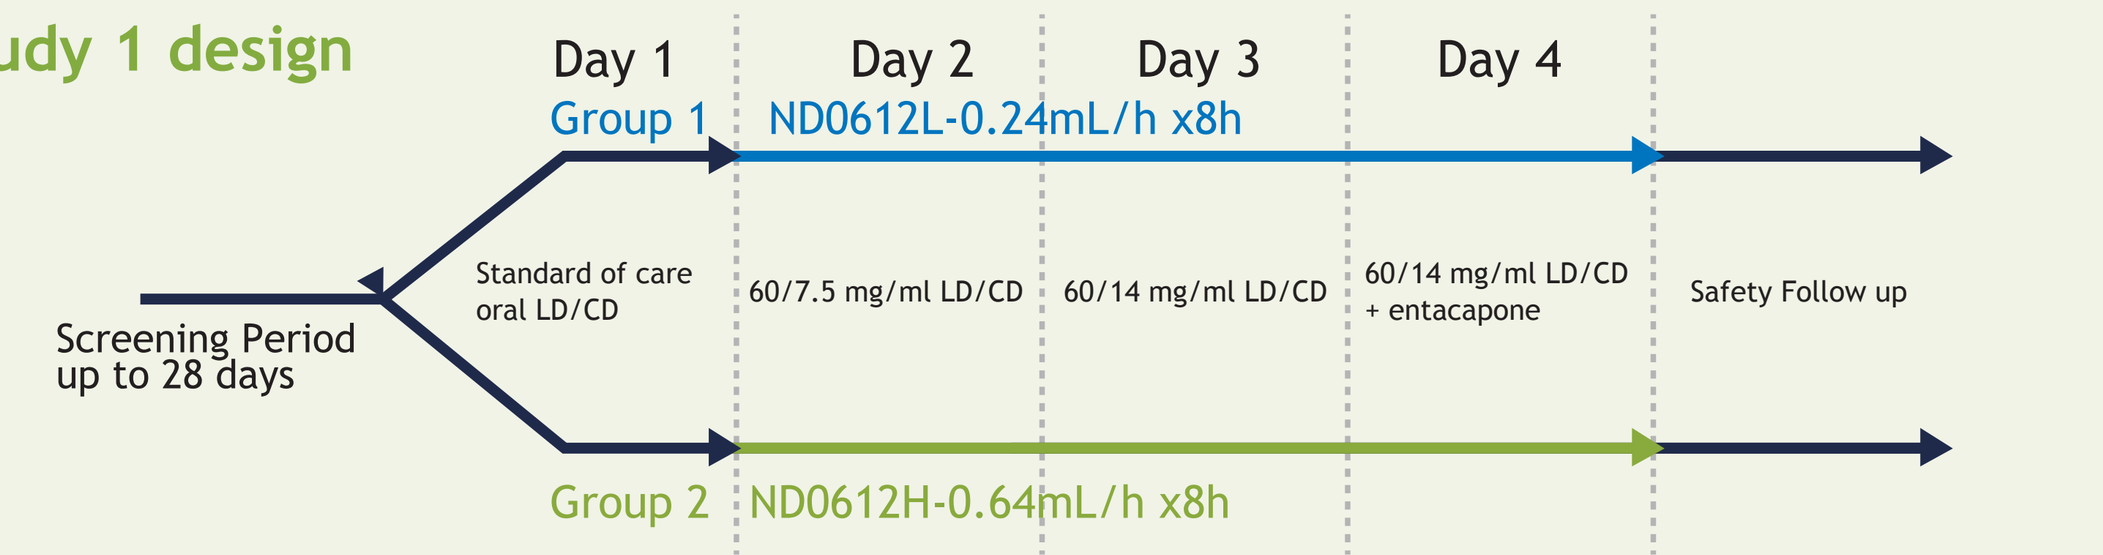

Day 1
Day 2
Day 3
Day 4
Group 1
ND0612L-0.24mL/h x8h
Screening Period
up to 28 days
Standard of care oral LD/CD
60/7.5 mg/ml LD/CD
60/14 mg/ml LD/CD
60/14 mg/ml LD/CD + entacapone
Safety Follow up
Group 2
ND0612H-0.64mL/h x8h

Day 1: all patients were treated with their standard oral levodopa.
Day 2: patients were treated with allocated regimen (ND0612L or ND0612H) administered with carbidopa at a concentration of 7.5mg/mL.
Day 3: patients received their allocated levodopa regimen with carbidopa at a concentration of 14mg/mL.
Day 4 (not relevant for this analysis) was an optional evaluation of the impact of dual enzyme inhibition (DDC and COMT inhibition) with carbidopa and entacapone.

Study 2: ND0612-005a

- This study characterized the impact of three descending doses of carbidopa (7.5, 6 and 4 mg/mL) on levodopa bioavailability.
- The study was conducted in clinic, and included 21 male or female healthy volunteers (40-65y).
- Subjects were allocated to 3 consecutive infusions of either ND0612H (over 30 hours to a total levodopa dose of 749mg) or ND0612L (over 24 hours to a total levodopa dose of 270mg) in a non-randomized manner (Figure 1). Each consecutive infusion was given with decreasing concentrations of carbidopa and there was a washout of ≥18h between infusions.

Study 2 design

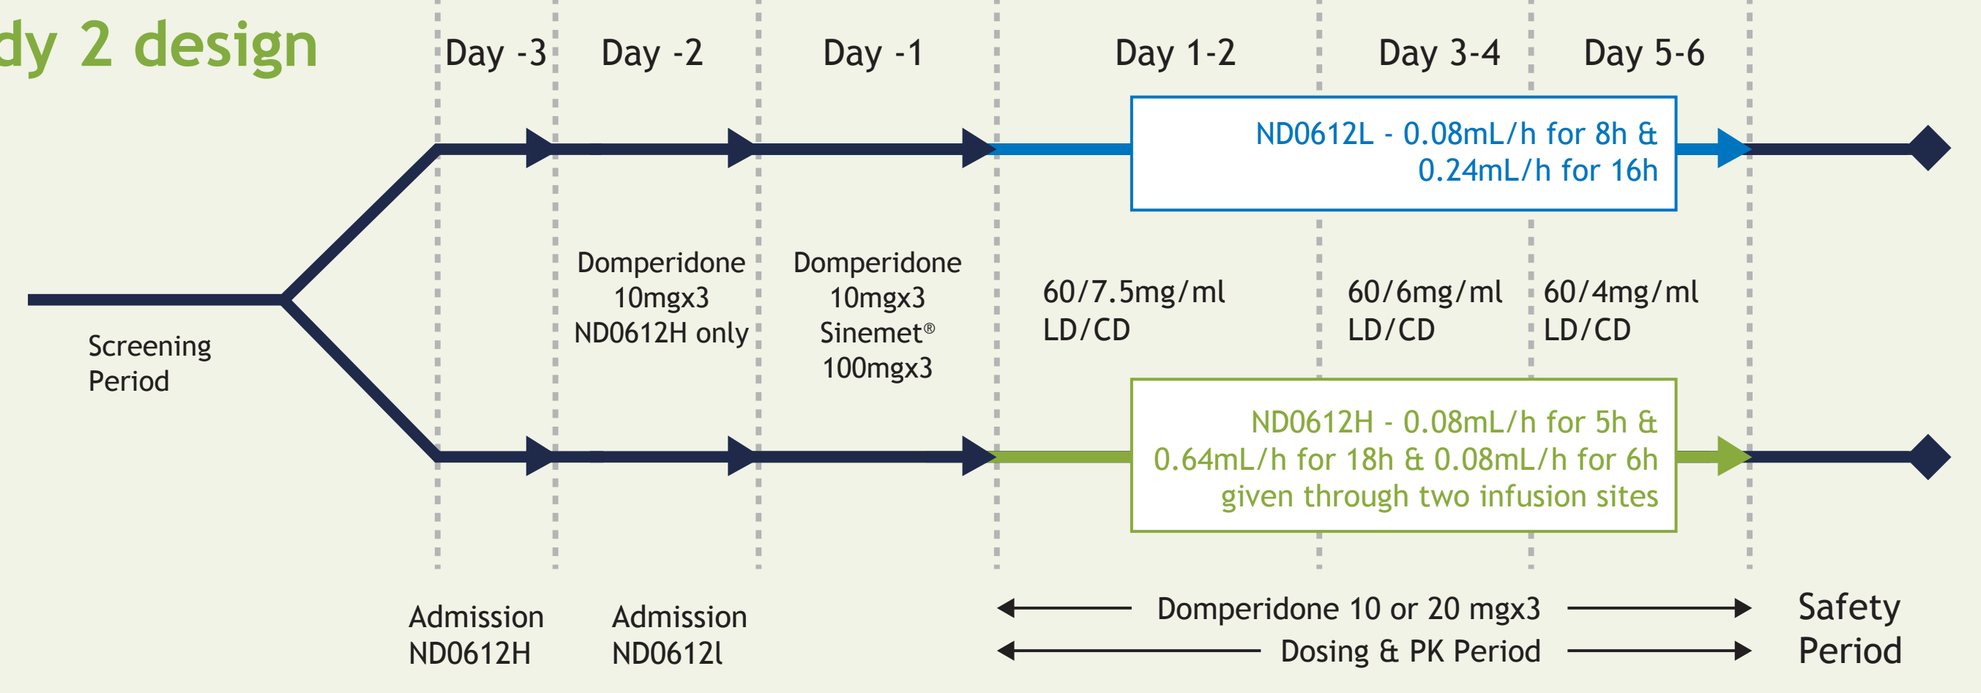

Day -3
Day -2
Day -1
Day 1-2
Day 3-4
Day 5-6
ND0612L - 0.08mL/h for 8h & 0.24mL/h for 16h
Screening Period
Domperidone 10mgx3 ND0612H only
Domperidone 10mgx3 Sinemet® 100mgx3
60/7.5mg/ml LD/CD
60/6mg/ml LD/CD
60/4mg/ml LD/CD
ND0612H - 0.08mL/h for 5h & 0.64mL/h for 18h & 0.08mL/h for 6h given through two infusion sites
Admission ND0612H
Admission ND0612l
Domperidone 10 or 20 mgx3
Dosing & PK Period
Safety Period

This study was conducted in clinic. Subjects allocated to ND0612H were admitted on Day -3 and received pretreatment with domperidone on Day -2. Subjects allocated to ND0612L were admitted on Day -2. On Day -1, subjects in both groups were pre-treated with oral carbidopa/levodopa (25/100mg) given every 8 hours (3 doses).
Day 1-2: Subjects received their allocated ND0612 regimen with carbidopa given at a concentration of 7.5 mg/mL
Day 3-4: Subjects received their allocated ND0612 regimen with carbidopa given at a concentration of 6 mg/mL
Day 5-6: Subjects received their allocated ND0612 regimen with carbidopa given at a concentration of 4 mg/mL

Pharmacokinetic analyses

- Blood samples were taken for pharmacokinetic analysis from pre-dose until 32 hours post-dose in healthy volunteers and until 8 hours in PD patients.
- Standard PK parameters (Cmax and AUC) were estimated for plasma carbidopa and plasma levodopa.
- In Study 1, the relative levodopa bioavailabilities (as assessed by geometric mean ratios [GMRs] of C_{max} and $AUC_{0\text{-}inf}$) of ND0612L and ND0612H at each carbidopa concentration treatment were compared using mixed modelling techniques. Study 1 also included exploratory trend analyses to investigate if there was an identifiable trend between the increasing dose of carbidopa for levodopa exposure levels for both ND0612L and ND0612H.
- In Study 2 the Fluctuation Index of levodopa, was assessed using a mixed measure repeated model (MMRM) analysis of Model's root mean square estimate (MSE). Data for ND0612L and ND0612H were separately compared versus the oral carbidopa/levodopa regimen, and for each carbidopa dose level.

Results

Study 1: ND0612 Pharmacokinetics in patients with PD

- Fluctuations in levodopa plasma levels were significantly reduced (<0.0001) for all ND0612 regimens versus oral levodopa/carbidopa.
- Both ND0612 regimens maintained steady, therapeutic levodopa plasma concentrations. Levodopa plasma concentrations were dose proportional with ND0612H achieving around 3-fold higher plasma levels than ND0612L, which is in accordance with the approximately 3-fold increase in daily levodopa dose infused in the ND0612H versus ND0612L treatment groups.
- No relevant differences in levodopa bioavailability were observed with the two carbidopa concentrations (Figure 1).
- Carbidopa exposure levels increased proportionally with carbidopa dose level (both ND0612 regimens).

Figure 1: Levodopa bioavailability with varying carbidopa doses (A) ND0612L (B) ND0612H

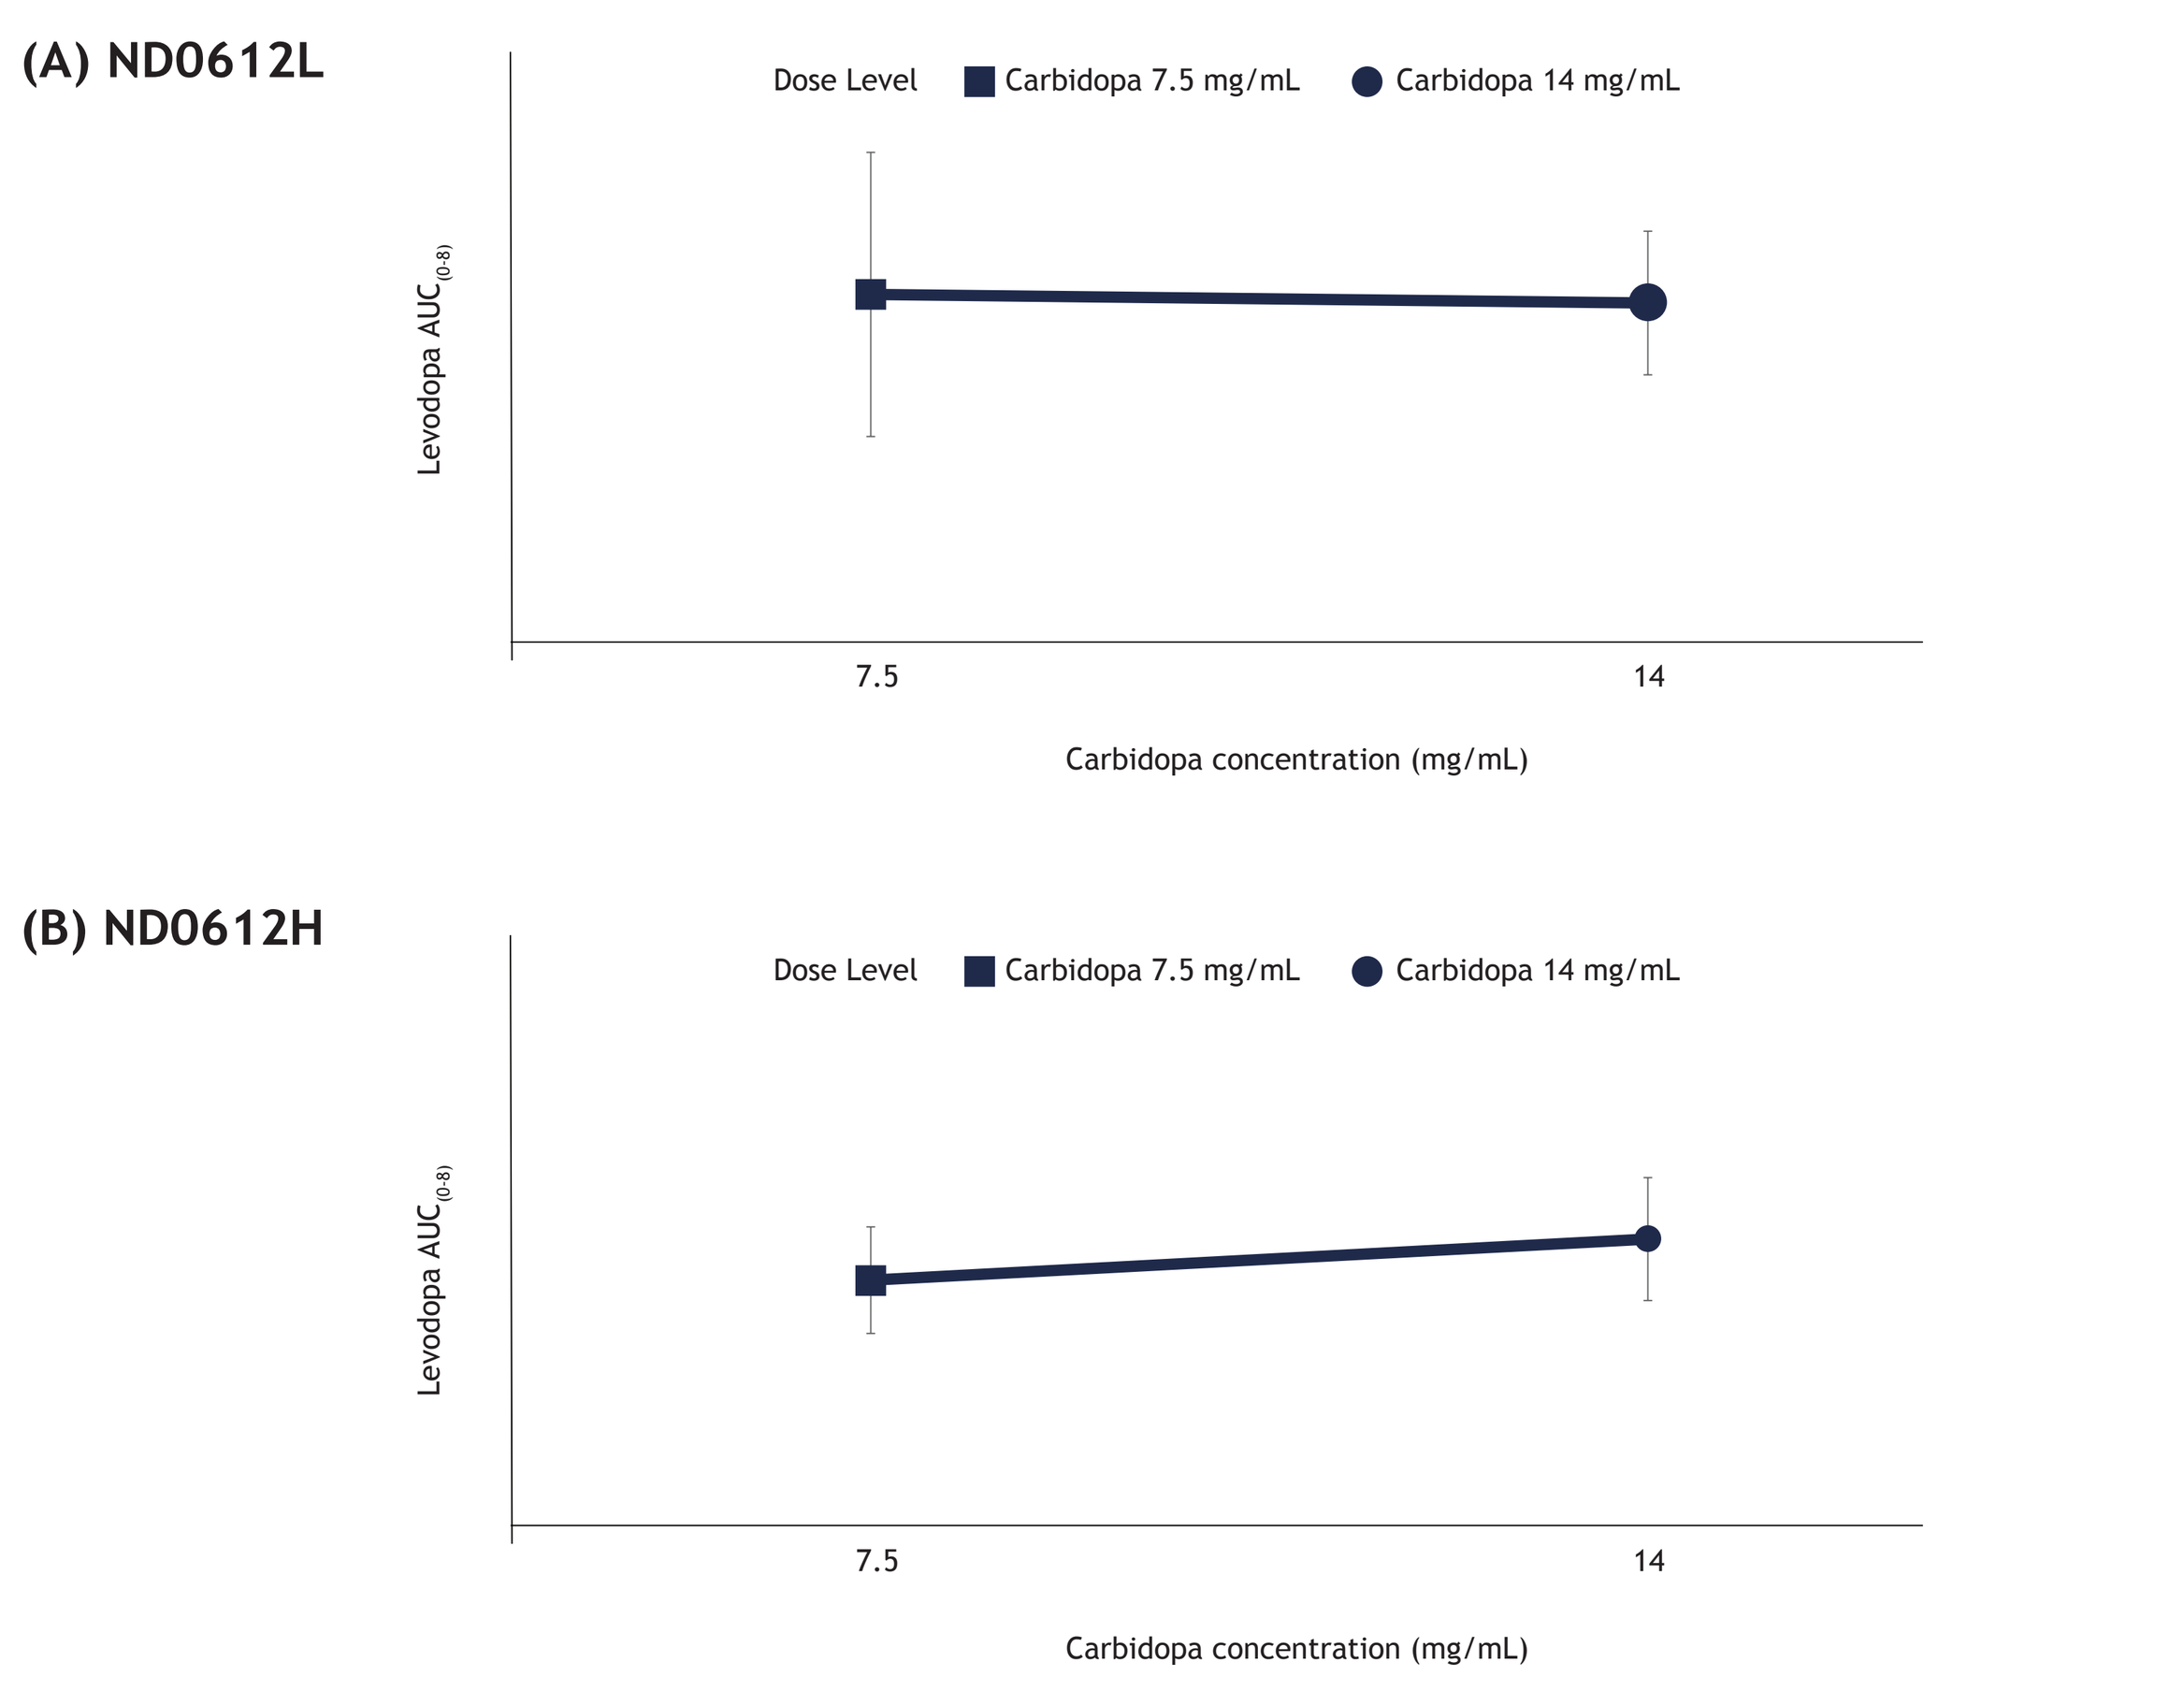

(A) ND0612L
Dose Level
Carbidopa 7.5 mg/mL
Carbidopa 14 mg/mL
Levodopa AUC(0-8)
7.5
14
Carbidopa concentration (mg/mL)
(B) ND0612H
Dose Level
Carbidopa 7.5 mg/mL
Carbidopa 14 mg/mL
Levodopa AUC(0-8)
7.5
14
Carbidopa concentration (mg/mL)

Figure 2: Levodopa bioavailability with varying carbidopa doses (A) ND0612L (B) ND0612H

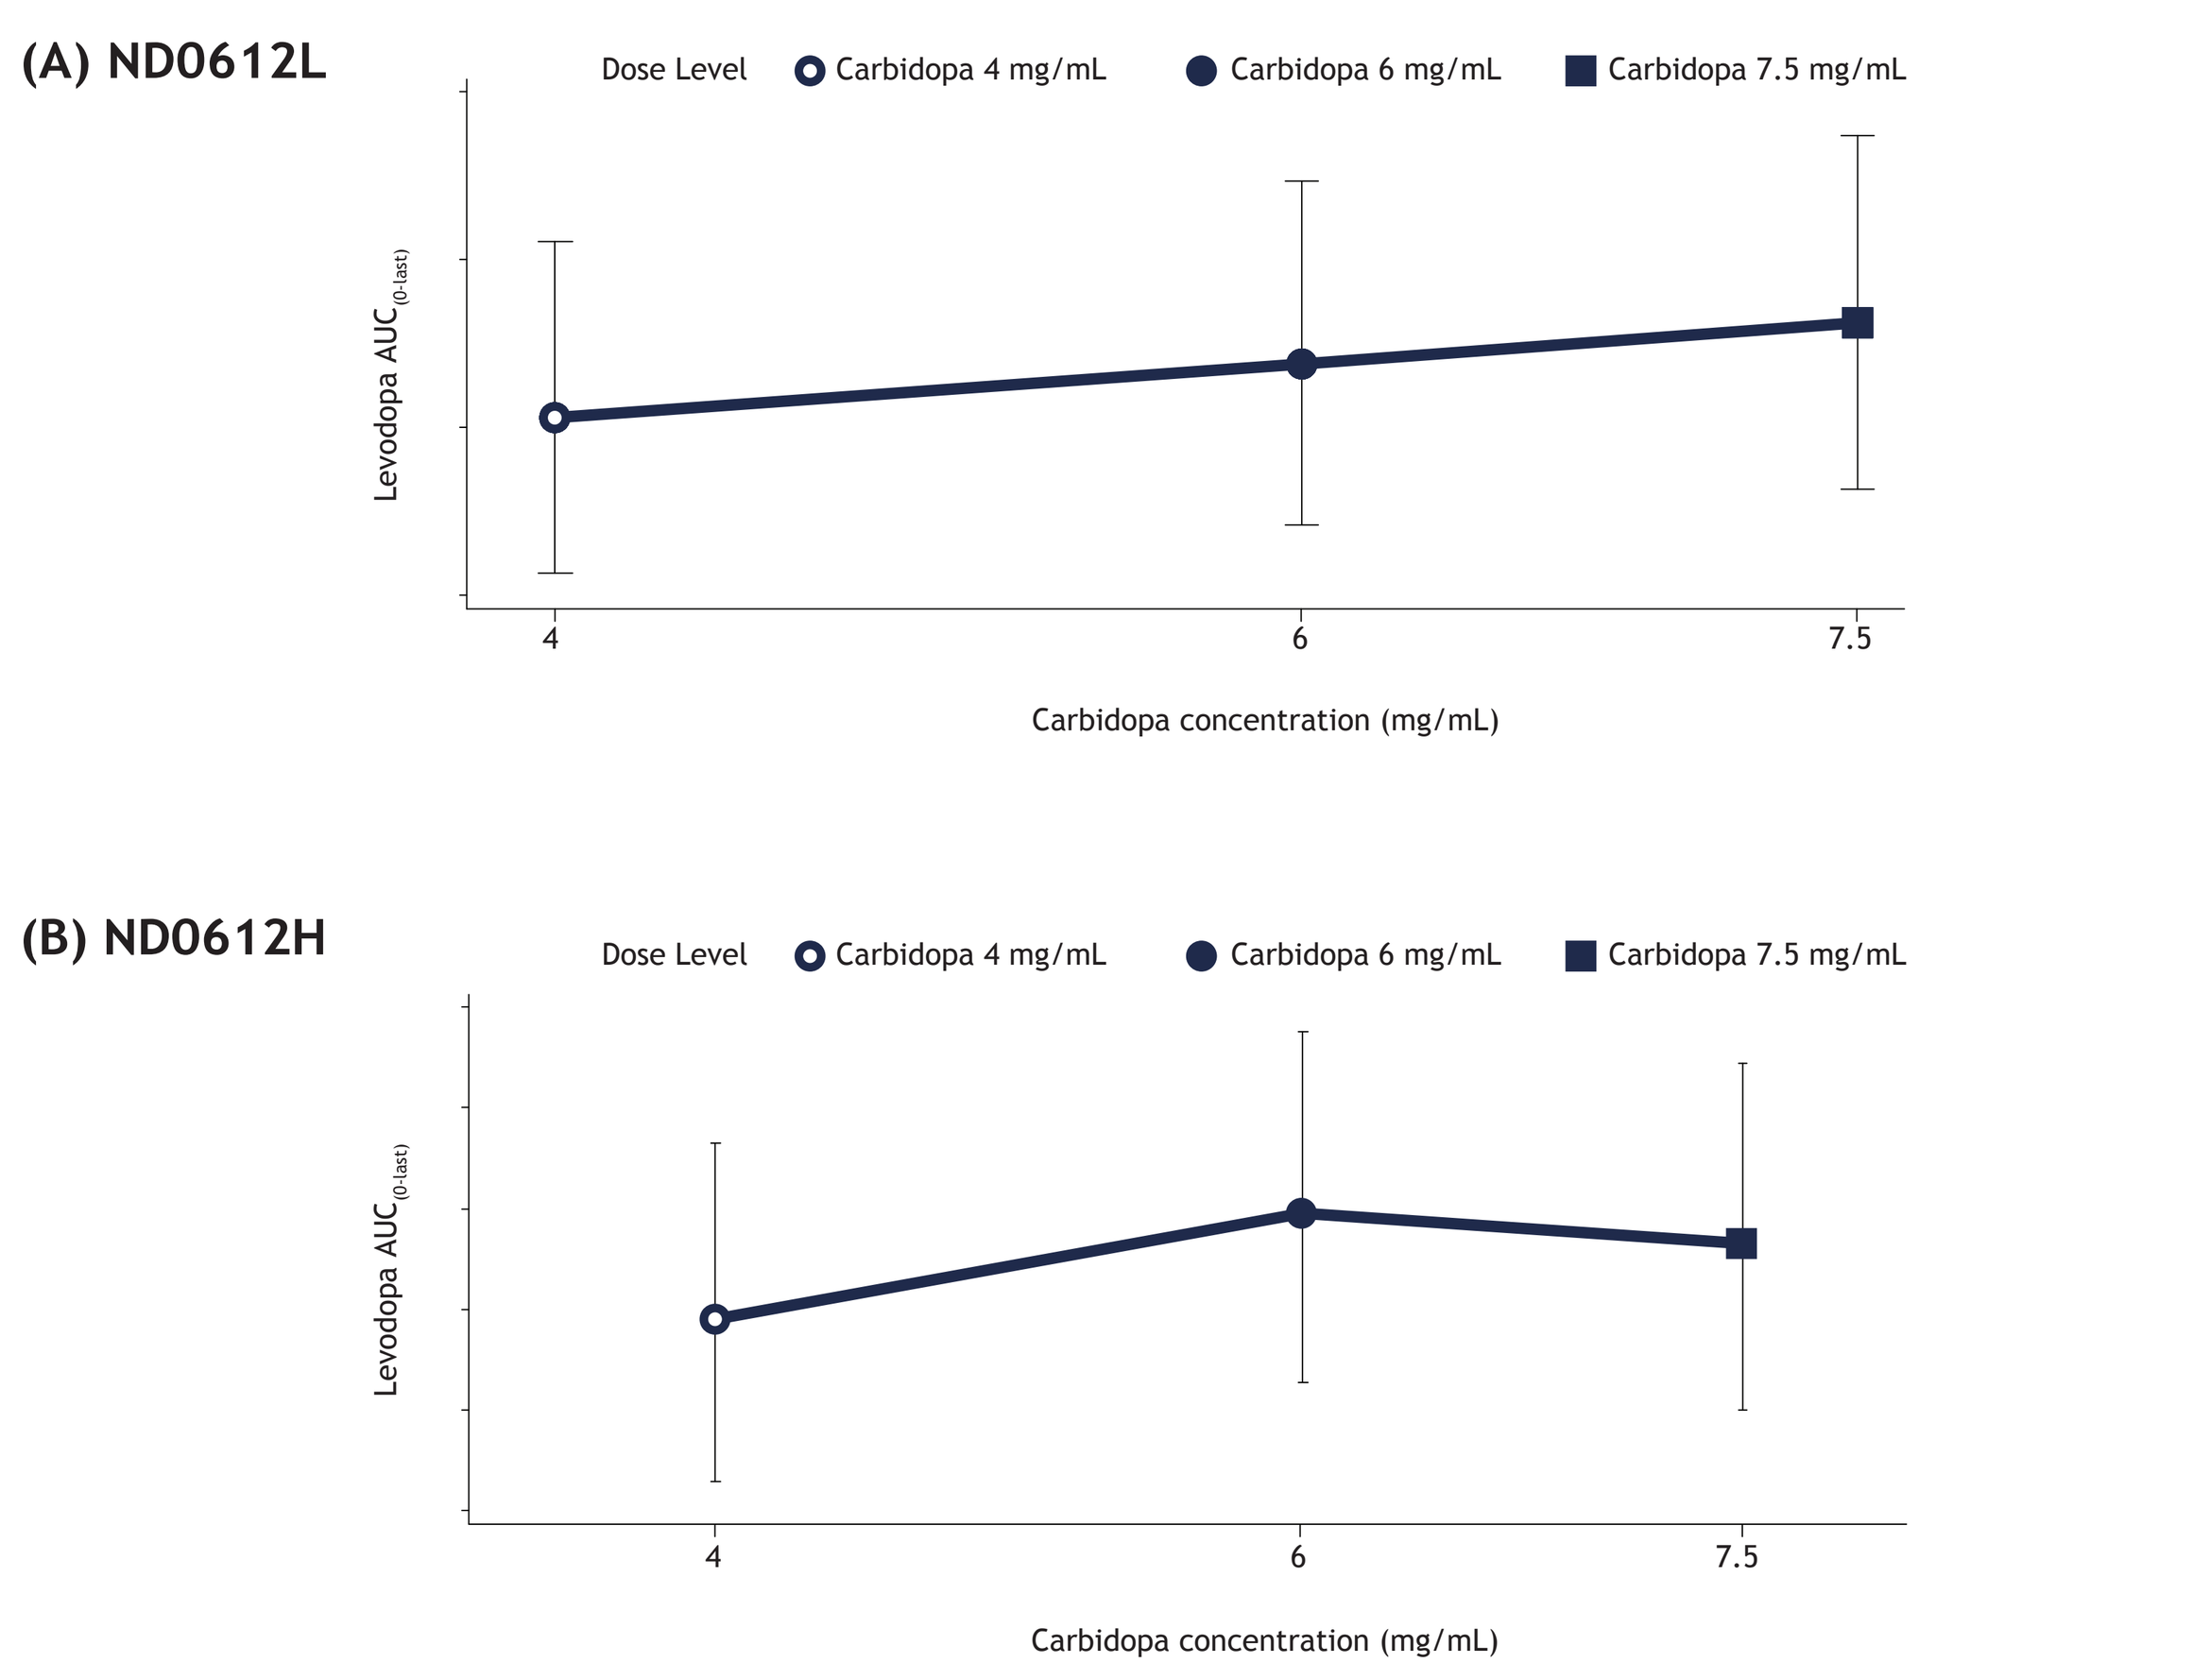

(A) ND0612L
Dose Level
Carbidopa 4 mg/mL
Carbidopa 6 mg/mL
Carbidopa 7.5 mg/mL
Levodopa AUC(0-last)
4
6
7.5
Carbidopa concentration (mg/mL)
(B) ND0612H
Dose Level
Carbidopa 4 mg/mL
Carbidopa 6 mg/mL
Carbidopa 7.5 mg/mL
Levodopa AUC(0-last)
4
6
7.5
Carbidopa concentration (mg/mL)

Study 2: ND0612 Pharmacokinetics in healthy volunteers

- Comparisons of GMRs indicated, that over the whole infusion period, observed levodopa exposure levels were similar (≤7% difference for the ND0612L regimen and ≤5% difference for the ND0612H regimen) between the three carbidopa dose levels.
- However, exploratory analyses indicated a positive trend between carbidopa dose levels used in the ND0612L regimen with respect to levodopa exposure, with higher AUCs observed at a carbidopa dose of 7.5 mg/mL. This trend was not apparent for the entire range of carbidopa doses when given with the ND0612H regimen (Figure 2).
- As expected, carbidopa exposure levels increased proportionally with carbidopa dose level (both ND0612 regimens). The 7.5 mg/mL dose of carbidopa consistently resulted in an AUC above 2000 ng.h/mL, which is considered to provide maximal inhibition of DDC.[4]

Conclusions:

- Taken together, these two studies confirmed that the concentration of subcutaneous carbidopa that provides optimal levodopa bioavailability in ND0612 is 7.5mg/mL.
- Increasing the carbidopa concentration above this level did not improve the levodopa bioavailability. However, lower concentrations could potentially compromise levodopa bioavailability for the lower dose regimen (ND0612L).
- This dosing ratio equates to a carbidopa: levodopa dose ratio of 1:8. The lower dose of carbidopa required versus current oral carbidopa/levodopa formulations reflects the significantly higher bioavailability of subcutaneously administered carbidopa.
- Based on these results the final formulation of ND0612 was defined as carbidopa 7.5 mg/mL/ levodopa 60 mg/mL. With this formulation, steady levodopa levels were consistently maintained in PD patients within the desired levodopa range.

References

1. Nutt JG, Woodward WR, Anderson JL. The effect of carbidopa on the pharmacokinetics of intravenously administered levodopa: the mechanism of action in the treatment of parkinsonism. Ann Neurol 1985;18(5):537-543.
2. Kaakkola S, Mannisto PT, Nissinen E, Vuorela A, Mantyla R. The effect of an increased ratio of carbidopa to levodopa on the pharmacokinetics of levodopa. Acta Neurol Scand 1985;72(4):385-391.
3. Shaltiel-Karyo R, Zawoznik E, Weinstock I, et al. Continuous subcutaneous delivery of solubilized carbidopa improves the pharmacokinetic profile of levodopa [abstract]. Mov Disord. 2016; 31 (suppl 2). http://www.mdsabstracts.org/abstract/continuous-subcutaneous-delivery-of-solubilized-carbidopa-improves-the-pharmacokinetic-profile-of-levodopa/. Accessed March 6, 2017.
4. Stalevo Prescribing Information. Novartis Pharmaceuticals Corporation. East Hanover, NJ. Last updated February 2016.

NeuroDerm