1377

Baseline characteristics of the population enrolled to a randomized clinical study of subcutaneous levodopa/carbidopa (ND0612) high dose infusion in patients with advanced PD

C Warren Olanow[1], Aaron Ellenbogen[2], Ruth Djaldetti[3], Tamar Rachmilewitz Minei[4], Yael Cohen[4], Sheila Oren[4], Karl Kieburtz[1]

[1]Clintrex, Rye, NY, USA, [2]Michigan Institute for Neurological Disorders and Quest Research Institute, Farmington, MI, USA, [3]Tel Aviv University, Tel Aviv, Israel [4]NeuroDerm Ltd., Rehovot, Israel

Introduction

- Continuous levodopa/carbidopa infusion is considered the optimal delivery route for treating patients with PD and advanced motor fluctuations because it avoids the peaks and troughs associated with oral dosing.[1, 2] However, current infusion systems must be surgically routed to the duodenum and are therefore associated with potentially serious complications.[3,4]
- NeuroDerm has developed ND0612, a proprietary novel liquid formulation of levodopa/carbidopa (60 mg/mL levodopa and 7.5 mg/mL of carbidopa) delivered by subcutaneous infusion via a belt pump system. ND0612 has been specifically developed to provide a continuous and stable source of levodopa in patients with moderate and advanced PD, while avoiding the surgical risks of intra-intestinal infusion.
- However, it is currently unknown if providing continuous 'round the clock' dopaminergic delivery (24 hours) with ND0612 offers any benefits over waking day infusions (e.g. reduced morning akinesia).
- This Phase II study aimed to evaluate whether continuous 24 hours delivery of subcutaneous ND0612 offers practical and clinical benefits over continuous delivery over the waking day (14 hours).

Methods

- ND0612-006 was an open-label, rater-blind, multicenter, parallel-group, randomized clinical study investigating the efficacy, pharmacokinetics, safety and tolerability of continuous subcutaneous infusion of two dosing regimens of high dose ND0612 compared to standard oral levodopa/DDI in PD patients experiencing motor fluctuations (NCT02577523).
- After completing 1 day of standard oral levodopa/carbidopa treatment, subjects were admitted as inpatients and randomized (1:1) to 28 days of treatment with ND0612 subcutaneous infusion administered for 24 hours (R1; total levodopa/carbidopa dose of 720/90 mg) or infusion during the waking day (R2; total levodopa/carbidopa dose of 538/67mg) infused over 14 hours with a morning bolus dose of oral levodopa/carbidopa.
- Patients in both regimens were allowed to take supplemental doses of oral levodopa/carbidopa as needed.
- Patients then continued as outpatients on a maintenance dose of the assigned ND0612 dosing regimen for the next 25 days.
- Patients and staff responsible for administering the study treatments were not blinded. However, all assessments of motor state were performed by a blinded rater.
- The primary endpoint was the change in OFF time from Baseline to Day 28 for each treatment regimen estimated using a Mixed Model for Repeated Measures (MMRM) including response data from Day 3 and 28 with no imputation for missing data.

Table 1. Key inclusion/exclusion criteria

Inclusion	Exclusion
Male and female (30 - 80 years) patients with a diagnosis of PD	Atypical parkinsonism
Hoehn & Yahr (during ON) of stage ≤3	Mini Mental State Examination (MMSE) score ≤26
≥2.5 hours of OFF time per day.	Prior neurosurgical procedure for PD
Predictable and well defined early morning OFF periods and a 'good' early morning response to levodopa.	Prior treatment with Carbidopa Levodopa Enteral Suspension (CLES)
Taking ≥4 levodopa doses per day (≥3 doses/day of IPX-066)	Any relevant medical, surgical, or psychiatric condition (including acute psychosis or hallucinations in prior 6 months), laboratory value, or concomitant medication which could compromise safety or potentially prevent completion of all aspects of the study.
Have attempted to take at least 2 other classes of PD medications in a therapeutic dose for at least 30 consecutive days each. All PD medications had to have been stable for ≥30 days prior to the start of the study.	

Figure 1: Study design

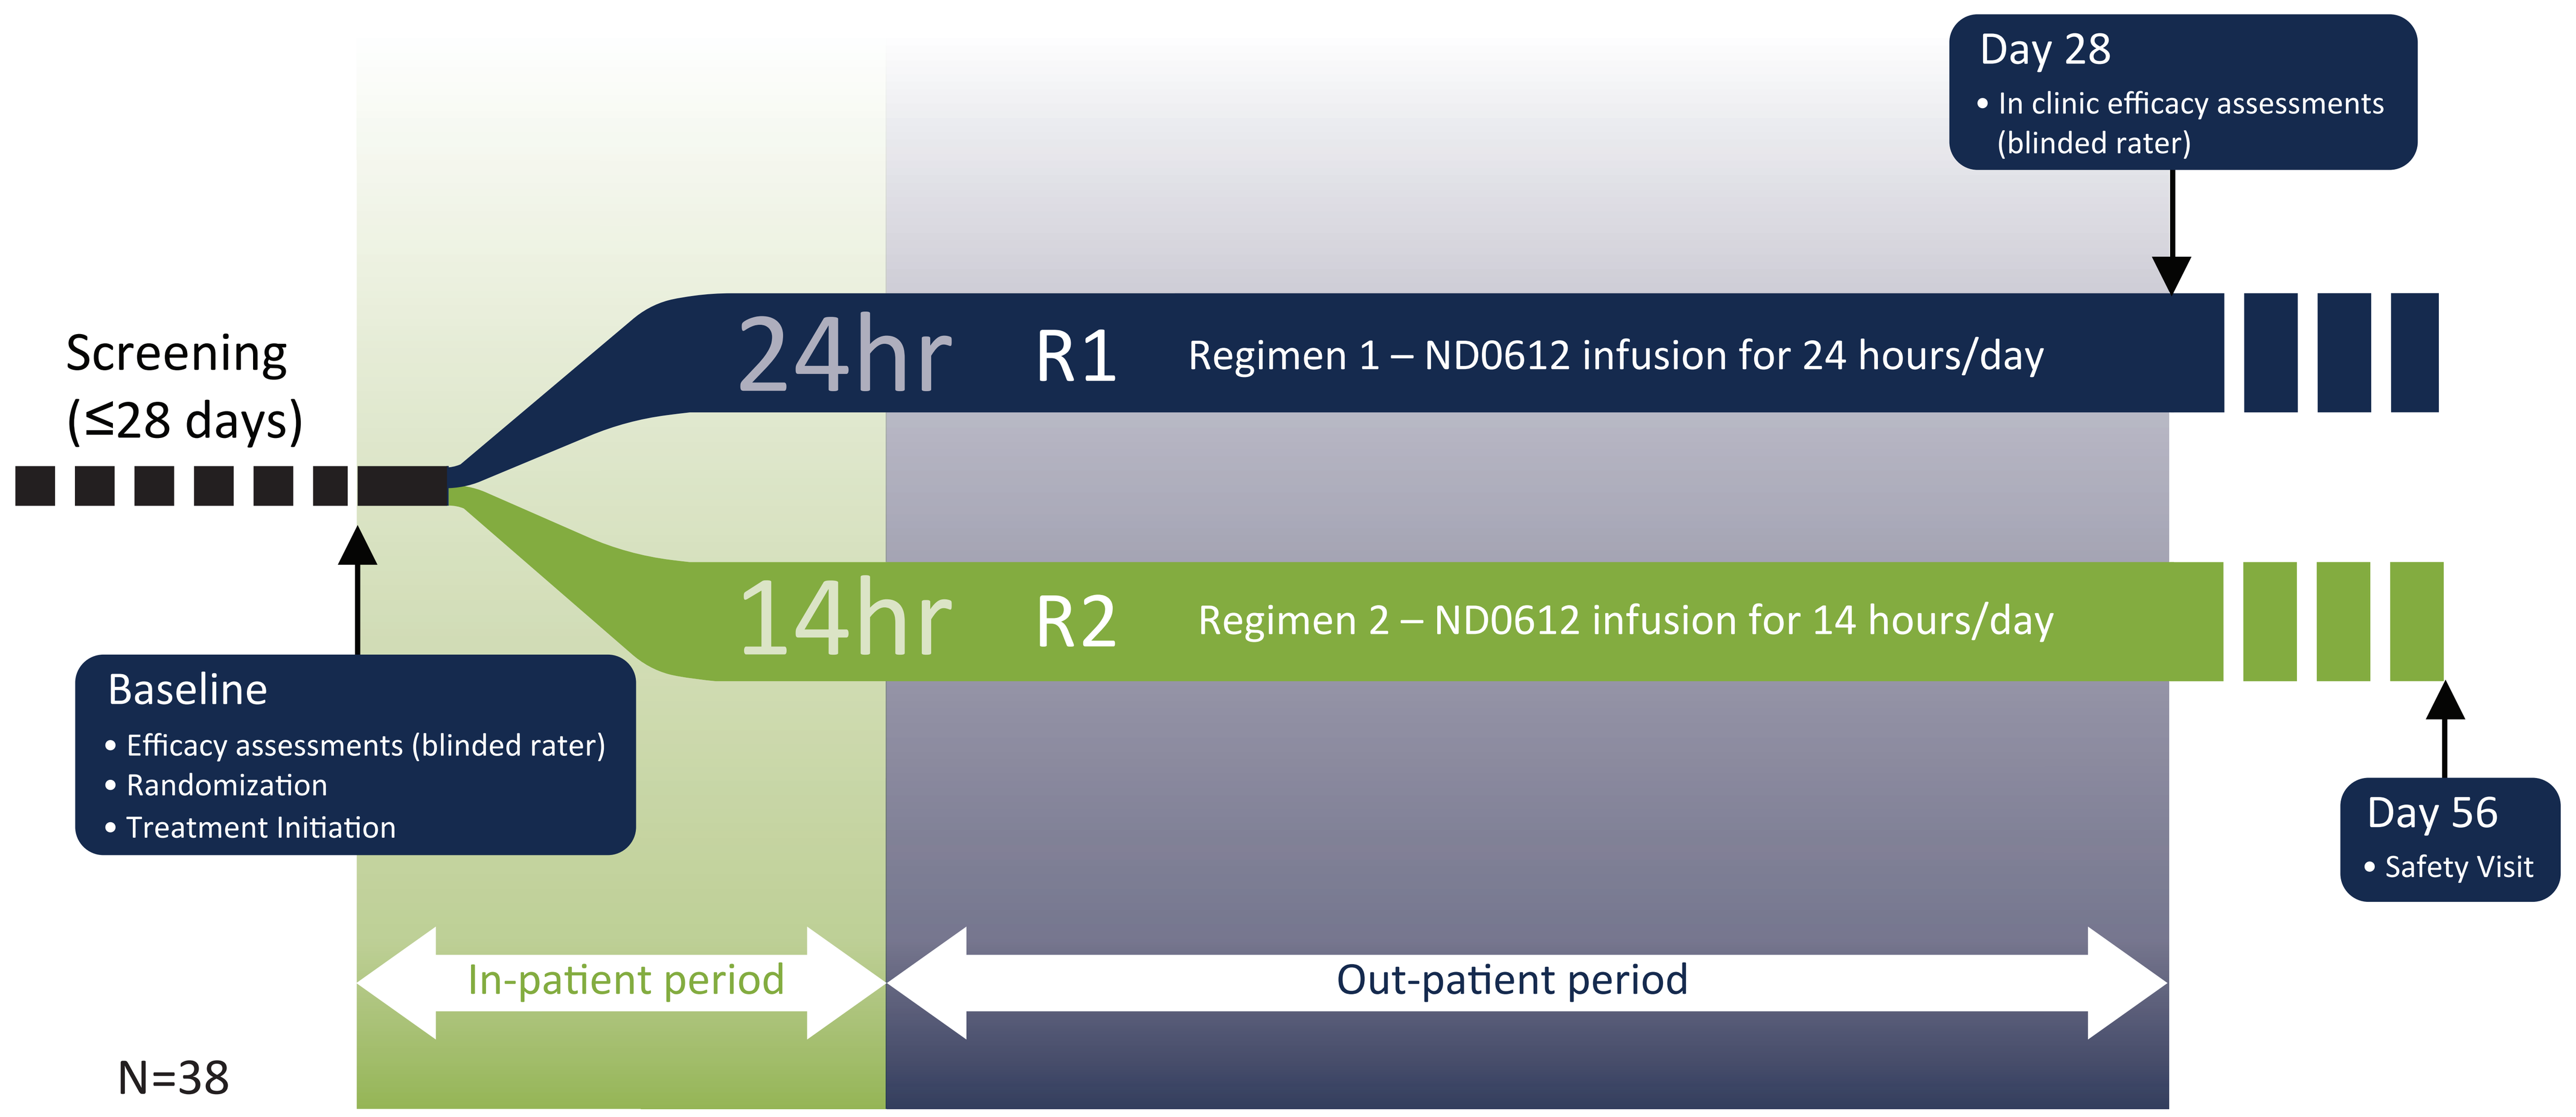
Day 28
• In clinic efficacy assessments (blinded rater)
Screening (≤28 days)
24hr R1 Regimen 1 – ND0612 infusion for 24 hours/day
14hr R2 Regimen 2 – ND0612 infusion for 14 hours/day
Baseline
• Efficacy assessments (blinded rater)
• Randomization
• Treatment Initiation
Day 56
• Safety Visit
In-patient period
Out-patient period
N=38

- Patients randomized to the 24-hours ND0612 group (R1) received a total dose of subcutaneous levodopa/carbidopa of 720/90 mg. Rates of infusion were higher in the daytime (18 hours; 0.64 mL/hr) and lower in the night (6 hours; 0.08 mL/hr). All subjects wore the pump during sleep.
- Patients randomized to the 14-hours ND0612 group (R2) received a total dose of subcutaneous levodopa/carbidopa dose of 537.6/ 67.2mg during the waking hours, and disconnected the pump during sleep. Patients also received a morning dose of oral levodopa/carbidopa (150/15 mg).

Table 2. Assessments

Efficacy	Safety/tolerability
• OFF time (primary) • Percentage of subjects with full ON at 8 am and 9 am (key secondary) • ON with and without dyskinesia • UPDRS (total, motor and activities of daily living) scores • Clinical Global Impression (Improvement and Severity) • Parkinson's Disease Sleep Scale • Need for dose adjustments of oral PD medication	• AE reporting • Local assessment of infusion sites (e.g. nodule formation, pain) • Daytime sleepiness (Epworth Sleepiness Scale) • Impulse control tolerability (Questionnaire for Impulsive-Compulsive Disorders in Parkinson's Disease-Rating Scale) • Suicidality (Columbia Suicide Severity Rating Scale) • Laboratory parameters, vital signs, physical and neurological examination • Electrocardiogram (ECG) evaluation
Patient reported outcomes	**PK**
• Subject's Global Impression of Improvement • Satisfaction with the dosing regimen • Night-time sleep	• Venous sampling for pharmacokinetic assessments on Days 2,3 and 28 (at 2 and 4 hours post dose)

Baseline

- 38 subjects were enrolled to the study from 11 sites in the United States (N=12), Europe (N=13) and Israel (N=13).
- Baseline characteristics are typical of patients with advanced PD who suffer from motor fluctuations despite optimized oral treatment. Baseline characteristics were comparable between the 2 groups.

Table 3: Baseline characteristics of the enrolled patients

	R1 (24 hours) (N=19)	R2 (14 hours) (N=19)	Overall (N=38)
Age (years); mean (SD)	63.0 (10.07)	64.0 (8.47)	63.5 (9.19)
Sex; n (%) male	12 (63.2)	14 (73.7)	26 (68.4)
Modified Hoehn and Yahr Stage; n (%)			
1	1 (5.3)	-	1 (2.6)
1.5	-	1 (5.3)	1 (2.6)
2	13 (68.4)	11 (57.9)	24 (63.2)
2.5	4 (21.1)	5 (26.3)	9 (23.7)
3	1 (5.3)	2 (10.5)	3 (7.9)
Years since PD diagnosis; mean (SD)	10.7 (5.5)	12.2 (5.0)	11.5 (5.2)
Years since motor fluctuations onset; mean (SD)	5.7 (6.9)	5.5 (4.8)	5.6 (5.9)
Years since dyskinesia onset	3.1 (2.7)	4.2 (3.4)	3.7 (3.1)
Daily OFF time (hours); mean (SD)	5.6 (2.1)	5.0 (2.4)	5.3 (2.2)
Daily Time with Moderate or Severe Dyskinesia; mean (SD)	1.2 (2.8)	2.5 (3.7)	1.9 (3.3)
UPDRS Part III (motor) score; mean (SD)	37.4 (14.5)	37.3 (13.3)	37.3 (13.7)
Levodopa dose (mg); mean (SD)	1135.8 (818.0)	1054.1 (567.3)	1094.9 (695.5)
Frequency levodopa dosing; mean (SD)	6.8 (3.2)	6.9 (2.2)	6.9 (2.7)

Late-breaking results

- In this Phase 2 study both regimens were well tolerated. The 24h infusion significantly reduced daily OFF time while increasing morning ON time and total daily good ON time. Please see Poster LBA41 for the late breaking efficacy results.

References

1. Olanow CW, Stern MB, Sethi K. The scientific and clinical basis for the treatment of Parkinson disease (2009). Neurology 2009;72(21 Suppl 4):S1-136.
2. LeWitt PA. Levodopa therapy for Parkinson's disease: Pharmacokinetics and pharmacodynamics. Mov Disord 2015;30(1):64-72.
3. Olanow CW, Kieburtz K, Odin P, et al. Continuous intrajejunal infusion of levodopa-carbidopa intestinal gel for patients with advanced Parkinson's disease: a randomised, controlled, double-blind, double-dummy study. Lancet Neurol 2014;13(2):141-149.
4. Antonini A, Odin P. Pros and cons of apomorphine and L-dopa continuous infusion in advanced Parkinson's disease. Parkinsonism Relat Disord. 2009;15 Suppl 4:S97-100.

NeuroDerm

1377

LBA41 (Late breaking results)