1337

Pharmacokinetic profile of continuous levodopa/carbidopa delivery when administered subcutaneously (ND0612) versus duodenal infusion (carbidopa and levodopa enteral suspension)

Liat Adar, Tamar Rachmilewitz Minei, Yael Cohen, Sheila Oren

NeuroDerm Ltd., Rehovot, Israel

Introduction

- The efficacy of continuous levodopa infusion in reducing motor complications in advanced PD patients is well established.[1]
- However, poor levodopa solubility has necessitated invasive gastrointestinal surgery to allow for direct infusion into the jejunum (site of levodopa absorption), thereby limiting the utility of current delivery systems such as the carbidopa/levodopa enteral suspension (CLES).[2]
- ND0612 is a proprietary liquid formulation (60 mg/mL levodopa/ 7.5 mg/mL carbidopa) delivered by an infusion pump system, specifically developed to enable subcutaneous (SC) administration. ND0612H is under development for use in severe PD as an alternative to surgical treatments.
- Previous studies have shown that treatment with ND0612 provides stable levodopa levels, and significantly reduces levodopa plasma level fluctuations in comparison to standard oral levodopa/carbidopa therapy (see Poster 1386 at this Congress).
- The aim of this study was to assess the pharmacokinetics and relative bioavailability of levodopa derived from ND0612 with CLES delivered via a naso-jejunal tube.

Methods

- This was an open-label, cross-over study conducted in 15 healthy subjects (aged 40-65 years).
- The study was performed in accordance with the Declaration of Helsinki and the International Conference on Harmonisation Good Clinical Practice guidelines. All subjects received a detailed explanation of the safety risks associated with direct naso-jejunal and subcutaneous levodopa delivery and provided written informed consent.
- Subjects were randomized (1:1:1) to 3 possible groups, each of which received 2 of 3 possible daily doses of CLES (washout of ≥24 hours between administrations), followed by SC ND0612 infusion (fixed dose) (Figure 1).
 - ND0612 was administered (using the belt dual syringe pump) over 24 hours; 6 hours were at a low infusion 'night' rate, which was then increased to a high 'day' infusion rate for 18 hours.
 - CLES, administration started with a morning bolus for 30 minutes followed by a constant naso-jejunal infusion for 15.5 hours.
- Venous blood samples were collected pre-dose and up to 24 hours or 32 hours post CLES administration and ND0612, respectively. Levodopa pharmacokinetic parameters ($C_{max,}$ AUC) derived from ND0612 and CLES and the relative bioavailability of levodopa from ND0612 to CLES (reference product) was evaluated.
- A formal statistical analysis was performed on the PK parameters AUC and C_{max} to assess relative bioavailability. Pharmacokinetic values were compared for (1) daily exposure and (2) for a truncated time interval (16 hours) representing 'waking' hours.
- Comparative bioavailability of levodopa between ND0612H and CLES was concluded if the 90% CI for adjusted geometric mean ratios (GMRs) lay within the acceptance limits of 80% to 125%.
- In addition, the safety and tolerability of each route of continuous administration was assessed.

Figure 1: Study design

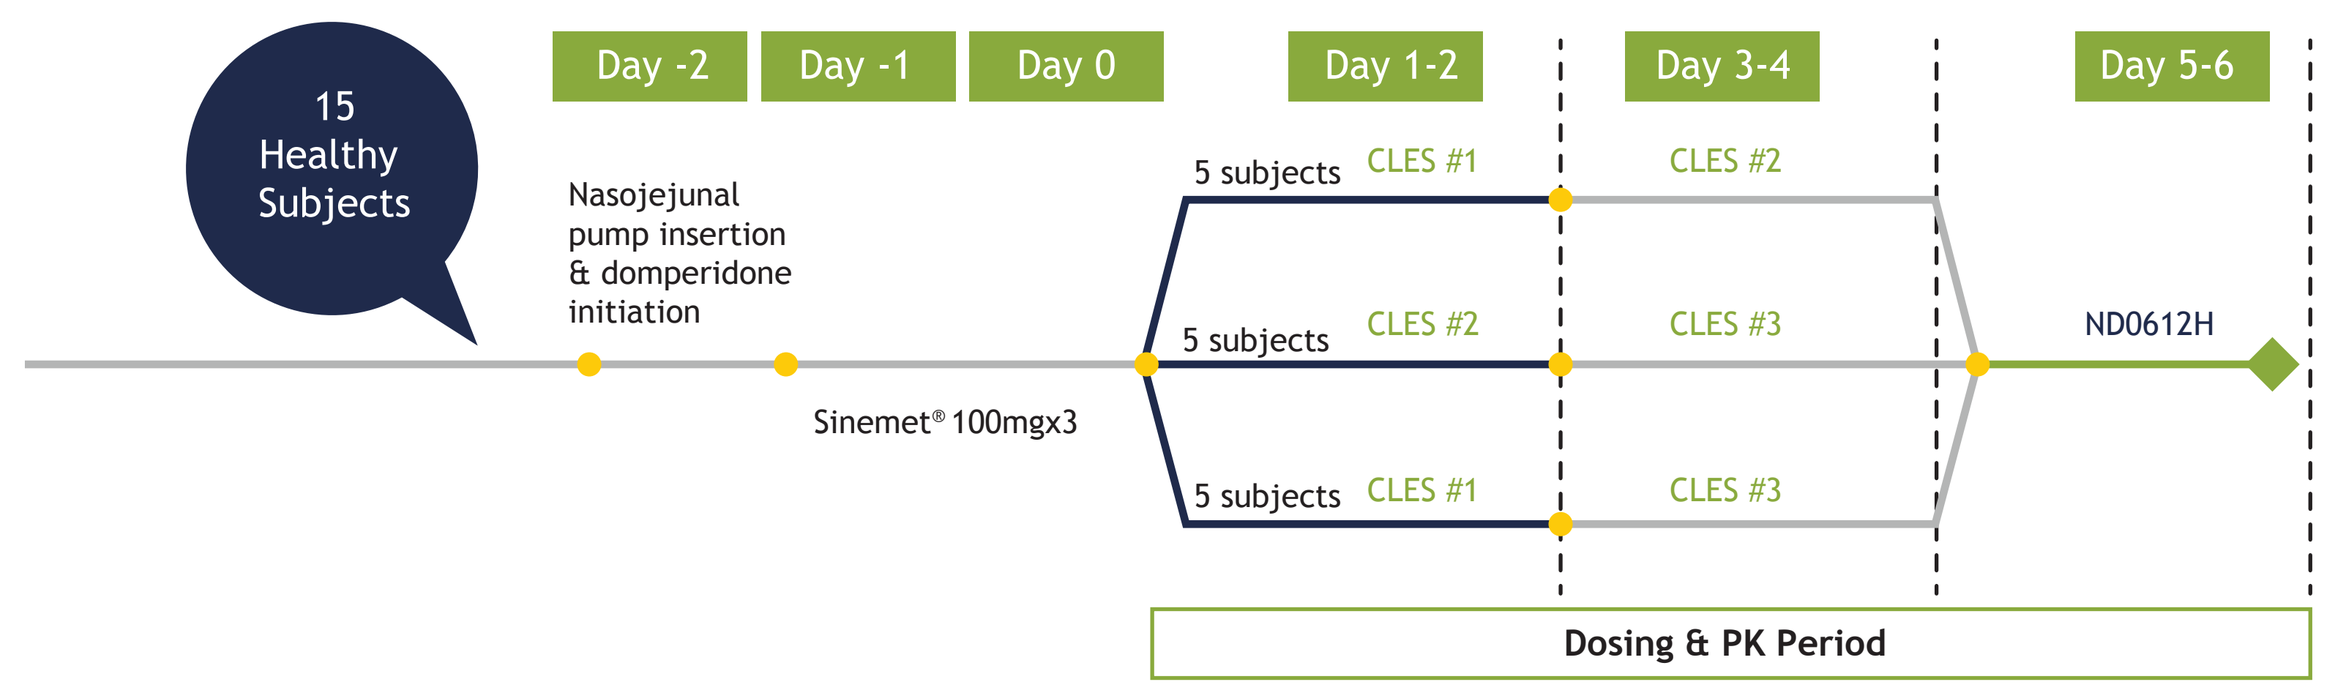

15 Healthy Subjects
Day -2
Day -1
Day 0
Day 1-2
Day 3-4
Day 5-6
Nasojejunal pump insertion & domperidone initiation
Sinemet® 100mgx3
5 subjects
CLES #1
CLES #2
5 subjects
CLES #2
CLES #3
5 subjects
CLES #1
CLES #3
ND0612H
Dosing & PK Period

In the evening of Day -2, a gastroenterologist fitted each subject with a naso-jejunal tube, which remained in place until completion of the last CLES dose. Subjects also received domperidone for the prophylactic treatment of possible levodopa gastrointestinal side effects from Day -2 to Day 6.

Figure 2: ND0612H achieved comparable pharmacokinetics to CLES within bioequivalence acceptance criteria
(a) overall plasma levodopa concentration time curves (b-d) plasma levodopa concentration time curves for individual subjects

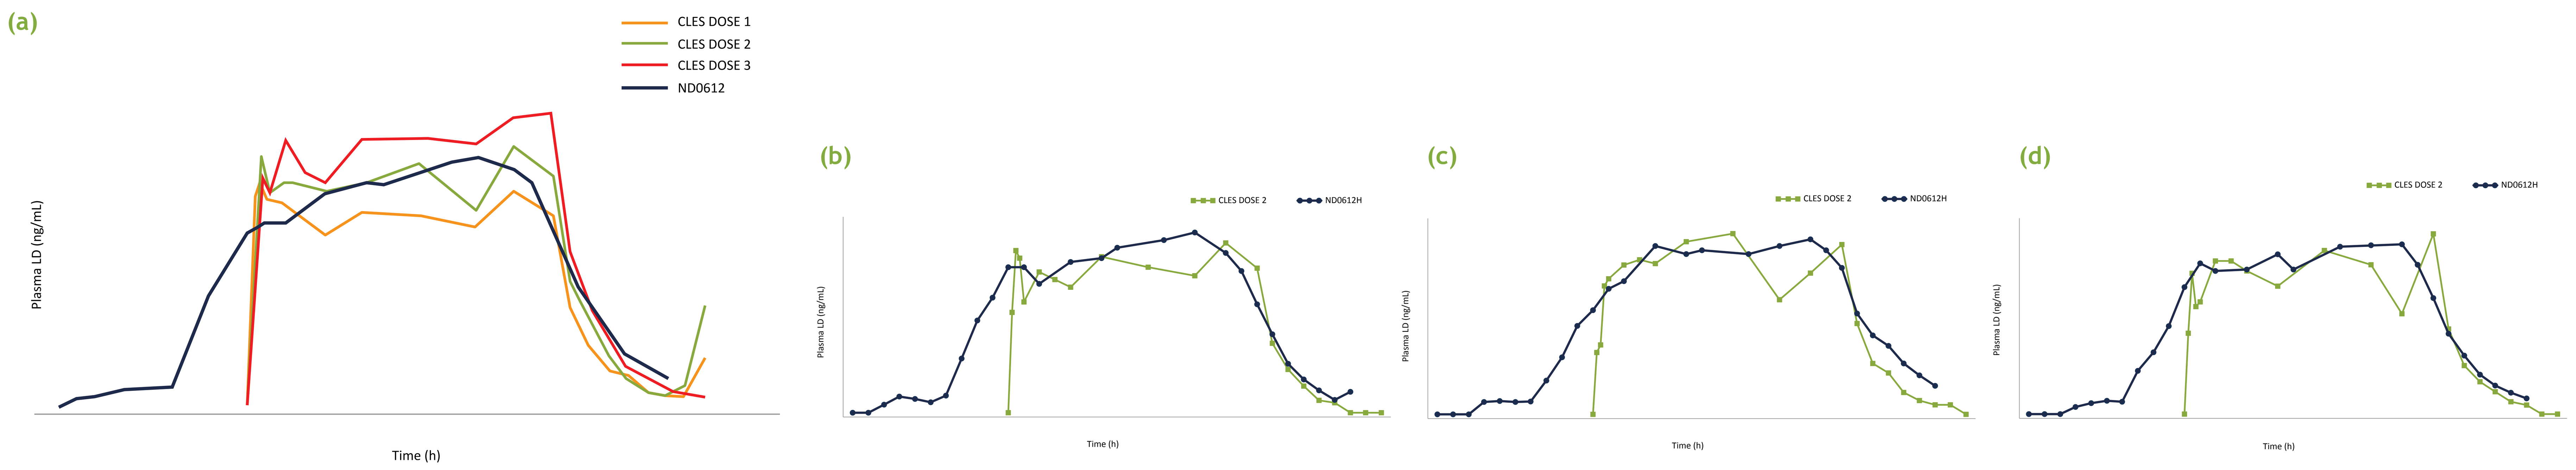

(a)
CLES DOSE 1
CLES DOSE 2
CLES DOSE 3
ND0612
Plasma LD (ng/mL)
Time (h)
(b)
CLES DOSE 2
ND0612H
Plasma LD (ng/mL)
Time (h)
(c)
CLES DOSE 2
ND0612H
Plasma LD (ng/mL)
Time (h)
(d)
CLES DOSE 2
ND0612H
Plasma LD (ng/mL)
Time (h)

Results

- A total of 15 subjects (14M/1F; mean ±SD age: 46.3 ±11.5 years) received study treatment. All 15 subjects completed the study and were included in the pharmacokinetic analysis (5 per arm).
- Administration of equivalent levodopa doses resulted in higher total plasma exposure (AUC) from ND0612 vs. CLES indicating a higher levodopa bioavailability from ND0612. In addition, intra- and inter-subject variability for ND0612 were overall lower compared with CLES.
- Figure 2 depicts the mean plasma levodopa concentration vs time profiles following nasogastric administration of the second dose of CLES and SC administration of ND0612H.
- When levodopa pharmacokinetic parameters for ND0612 were compared against CLES, the 90% confidence interval of the AUC and C_{max} adjusted geometric mean ratios (ND0612 to CLES) were within the range of 80-125% with adjusted GMRs close to 100% indicating bioequivalence.
- No new safety concerns were identified for ND0612. No severe or serious AEs were reported for any subject and no subject was withdrawn because of an AE.
- The most common AEs with ND0612 were site related and included injection site pain (n=7; 46.7%), induration (n=5, 33.3%), erythema (n=4, 26.7%) and swelling (n=3, 20.0%). The most common AEs with CLES were ganstrointestinal disorders including nausea (n=5, 33%) and diarrhea (n=2, 13.3%). were considered to be related to naso-jejunal administration.

Conclusions

- The 90% CI of the adjusted GMRs (AUC and Cmax) for ND0612 and all CLES doses were within the range of 80-125% indicating bioequivalence.
- Subcutaneous delivery of ND0612 provides higher levodopa bioavailability as compared to equivalent CLES dose.
- Levodopa levels derived from ND0612 demonstrates lower intra- and inter-subject variability compared to CLES.
- Subcutaneous delivery of levodopa/carbidopa may provide a safer alternative to direct intestinal infusion whilst providing clinical benefit to PD patients with motor fluctuations.
- This Phase II study is one of the first to evaluate the pharmacokinetics of direct naso-jejunal levodopa delivery in healthy volunteers, thereby permitting a direct comparison of CLES with ND0612. Both types of infusion were well tolerated.

References

1. Olanow CW, Obeso JA, Stocchi F. Continuous dopamine-receptor treatment of Parkinson's disease: scientific rationale and clinical implications. Lancet Neurol 2006;5(8):677-687.
2. Antonini A, Odin P. Pros and cons of apomorphine and L-dopa continuous infusion in advanced Parkinson's disease. Parkinsonism Relat Disord. 2009;15 Suppl 4:S97-100.

NeuroDerm