Pharmacokinetic profile of low-dose ND0612 (levodopa/carbidopa for subcutaneous infusion) in patients with moderate to severe Parkinson's disease

Nir Giladi[1], Yoseph Caraco[2], Tanya Gurevich[3], Ruth Djaldetti[4], Yael Cohen[5], Oron Yacobi-Zeevi[5] and Sheila Oren[5]

[1]Tel Aviv Medical Center, Tel Aviv University, Tel Aviv, Israel; [2]Hadassah Medical Center, Jerusalem, Israel; [3]Sackler School of Medicine, Tel Aviv University, Tel Aviv, Israel; [4]Tel Aviv University, Tel Aviv, Israel and [5]NeuroDerm Ltd., Rehovot, Israel.

Introduction

The symptomatic efficacy of continuous levodopa/carbidopa (LD/CD) delivery in PD patients with motor fluctuations is well known. Poor LD solubility has previously prevented development of a subcutaneously deliverable LD/CD formulation, therefore the currently available systems for continuous LD delivery have to be surgically inserted into the jejunum, and are associated with tolerability concerns. ND0612 is a proprietary liquid formulation of LD/CD that enables for the first time continuous subcutaneous infusion administration of LD/CD.

Study Objectives

To characterize the pharmacokinetic profile, and the safety and tolerability of continuous subcutaneous (SC) delivery of ND0612 in patients with advanced PD

ND0612L (LD/CD 60/14 mg/mL)

- 0.08 ml/h during 8 hours (low night rate, 18:00-2:00)
- 0.24 ml/h during the next 16 hours (high day rate, 2:00-18:00)
- Total daily volume: 4.5 ml
- Total LD daily dose: 270 mg
- Total CD daily dose: 63 mg

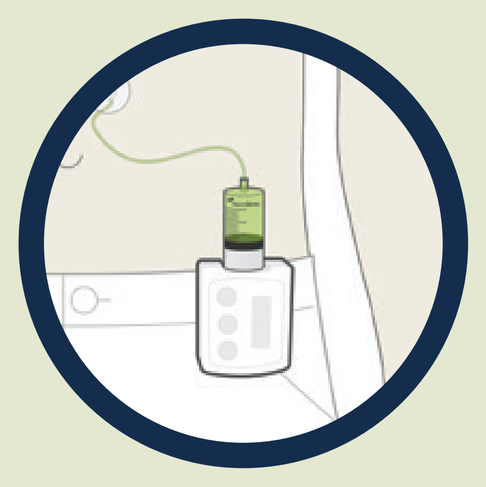

Methods

This was a Phase II randomized, placebo-controlled, double-blind, two-period study of low-dose ND0612 in patients with PD and motor fluctuations (Figure 1).

Figure 1: Study design

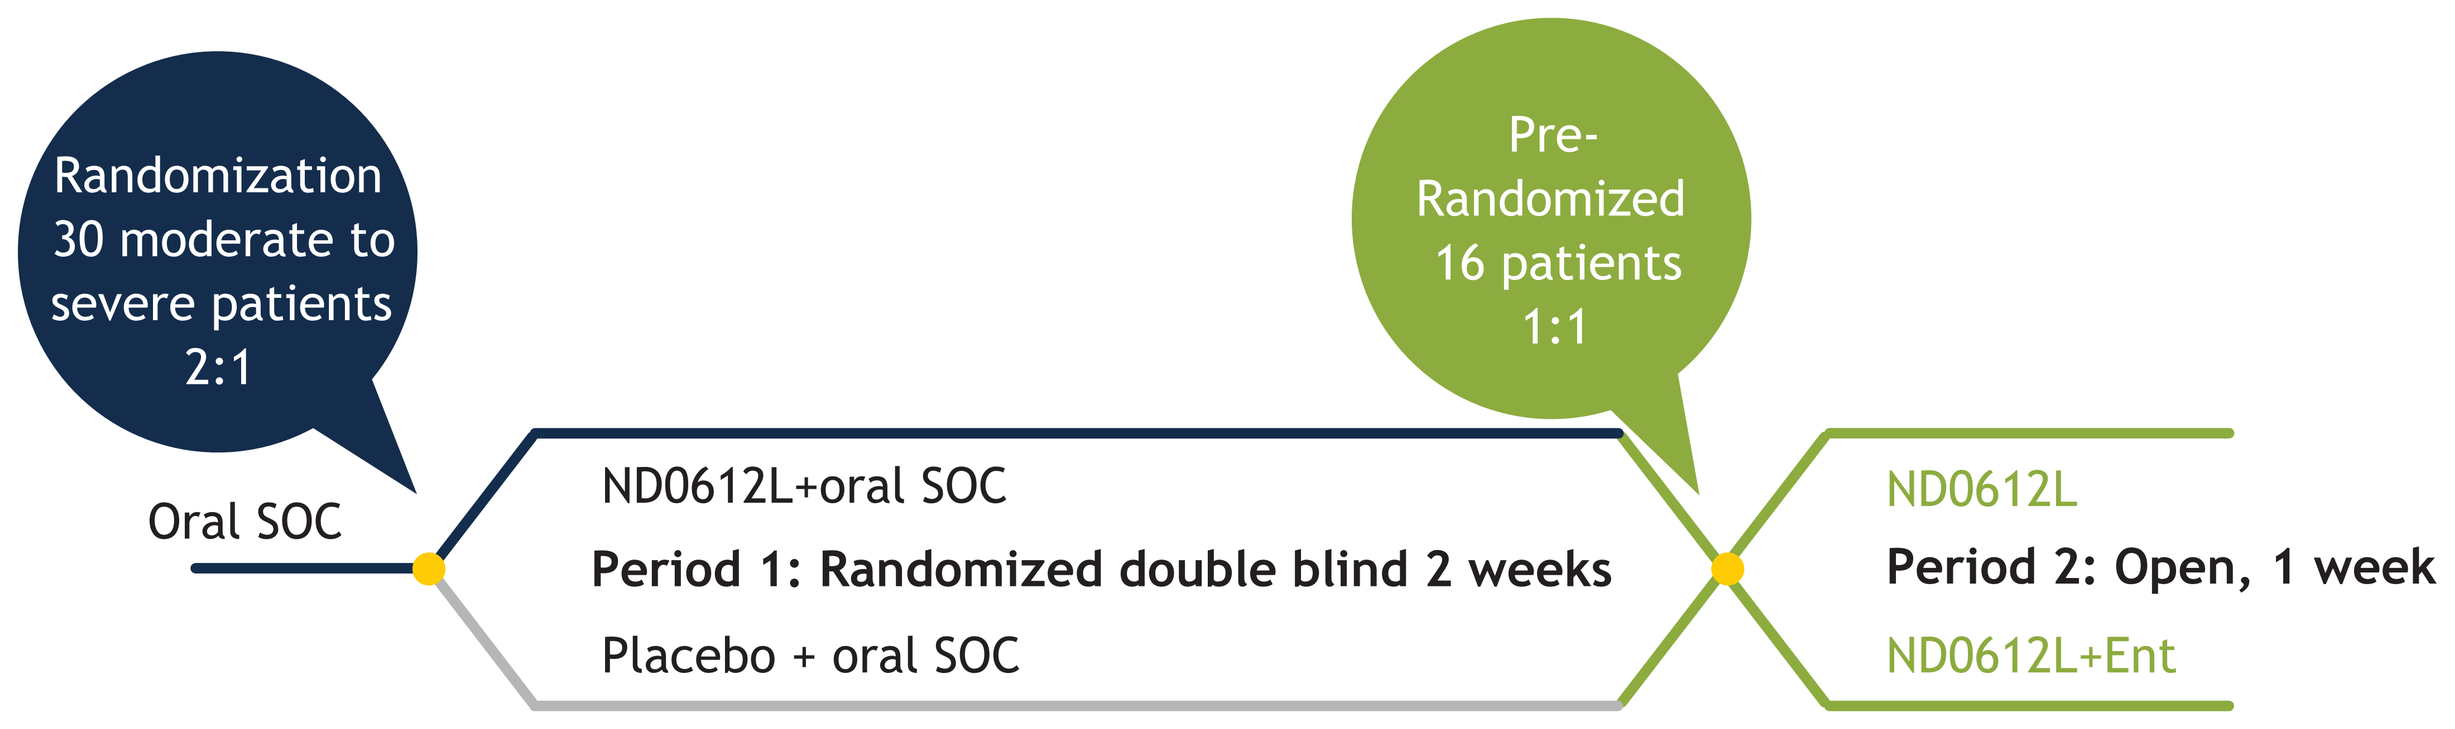
Randomization 30 moderate to severe patients 2:1
Oral SOC
ND0612L+oral SOC
Period 1: Randomized double blind 2 weeks
Placebo + oral SOC
Pre-Randomized 16 patients 1:1
ND0612L
Period 2: Open, 1 week
ND0612L+Ent

In Period 1

30 patients were randomized (2:1) to receive on top of their standard of care (SOC) oral LD/CD adjunct treatment with low-dose ND0612 or placebo for 14 days.

In Period 2

16 patients per protocol were treated for 7 days with open-label ND0612. The patients were randomized on a 1:1 ratio to 2 treatment arms: with additional entacapone 200mgX3, or without entacapone. Oral LD/CD was to be completely stopped unless needed by the patient.

Results

The study population was a typical representation of patients with moderate-severe advanced PD. The patients' baseline demographic and disease characteristics are presented in Table 1.

Table 1: Baseline characteristics

	ND0612L N=19	Placebo N=11
Age (years)	63.8 ± 7.4	64.5 ± 6.9
PD duration (years)	8.6 ± 4.5	8.7 ± 3.8
Duration of LD therapy (years)	7.1 ± 4.2	7.5 ± 3.3
Duration of fluctuations (years)	5.7 ± 4.2	5.5 ± 2.9
Duration of dyskinesias (years)	2.9 ± 2.0	3.4 ± 1.8
Total daily LD dose (mg)	659.9 ± 428.2	743.2 ± 355.0
Daily home OFF time (hrs)	5.85 ± 2.83	5.67 ± 2.73
Daily in-clinic OFF time (hrs)	5.92 ±2.14	4.64 ± 2.89
Daily home ON time with troublesome dyskinesia (hrs)	0.92 ± 1.60	1.23 ± 2.37
Daily in-clinic ON time with troublesome dyskinesia (hrs)	0.67 ± 1.45	0.14 ± 0.45

Pharmacokinetics

Patients treated with adjunct low-dose ND0612 had a markedly lower peak-to-trough ratio, a lower fluctuation index (FI) and lower coefficient of variation (CV) (Table 2).

Moreover, the deep LD plasma concentration troughs were eliminated in the ND0612 treated patients compared to baseline or placebo treated patients and their Cmax did not increase. (Figure 2). The number of hours LD plasma concentrations were above 1000 ng/mL increased by 4.4 (± 2.2) hours in the ND0612 treatment group compared to the placebo treatment arm.

In the 16 patients in period-2, intake of oral daily levodopa was markedly reduced by a mean of 516 [±486] mg. Nine out of 16 patients did not receive any oral LD on the day of PK sampling. In this subgroup of 9 patients, the LD concentration peaks observed with oral LD therapy were eliminated, resulting in a remarkably stable concentration of LD in the plasma without peaks or troughs (Figure 3). Plasma LD levels were maintained at a mean of 550±79ng/ml without entacapone and 800 ±144ng/ml with oral entacapone.

Table 2: Mean (±SD) Fluctuation index (FI) and Coefficient of Variation (CV)

	ND0612 (N=19)			Placebo (N=11)		
	Baseline	Day 14	Change from BL to D14	Baseline	Day 14	Change from BL to D14
FI	2.81 (±1.09)	1.61 (0.45)	**-1.19 (±1.00)**	3.11 (±1.08)	3.11 (±1.59)	**0.01 (±1.45)**
CV	0.85(±0.27)	0.48(0.11)	**-0.37(±0.26)**	0.90(±0.29)	0.92(±0.43)	**0.02(±0.30)**

Figure 2: ND0612L transforms levodopa pharmacokinetics

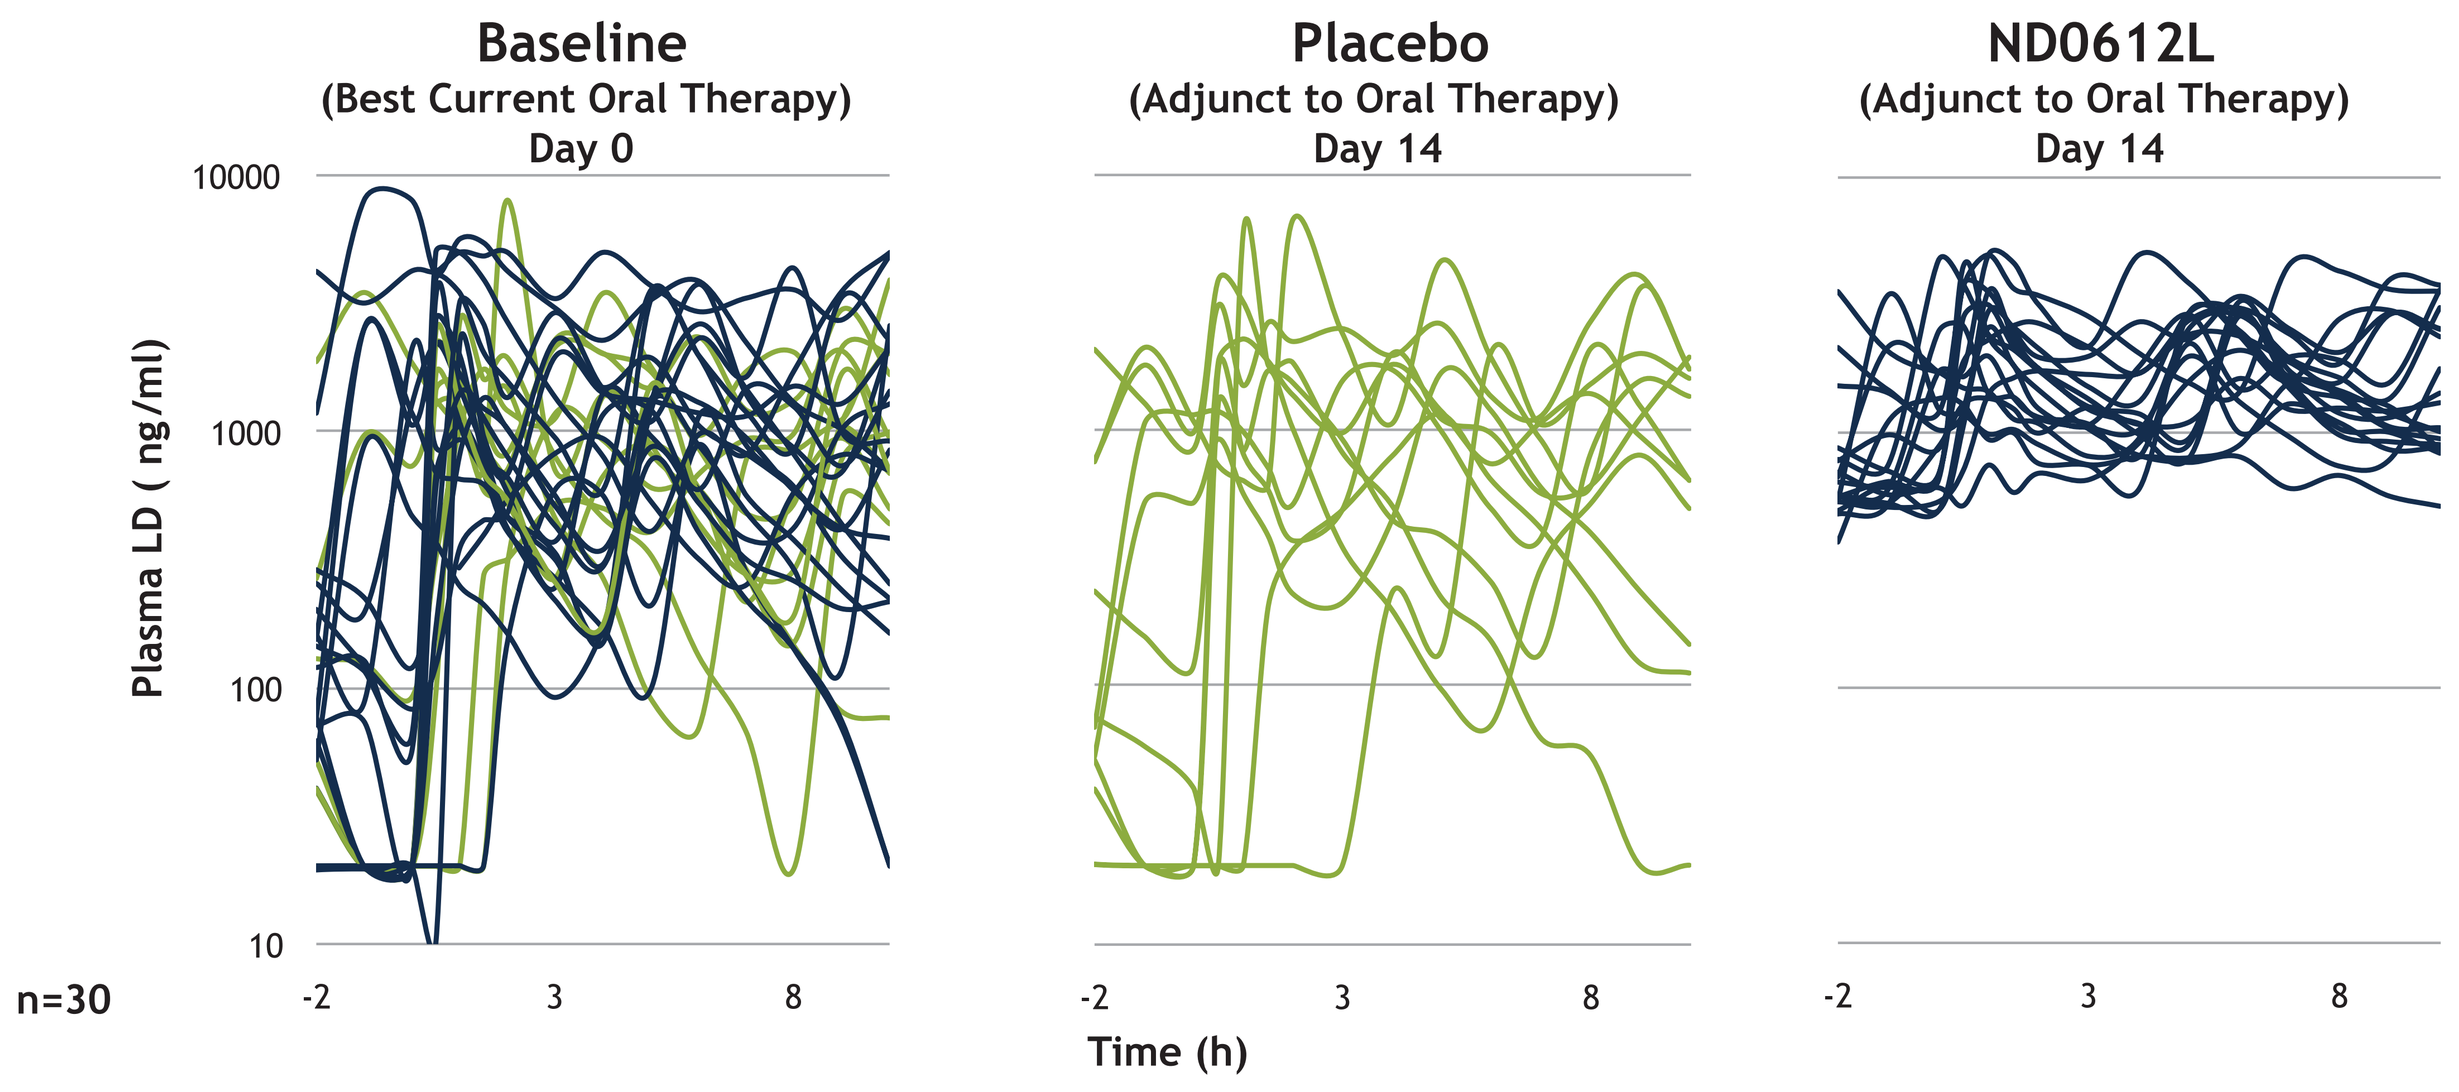
Baseline
(Best Current Oral Therapy)
Day 0
Placebo
(Adjunct to Oral Therapy)
Day 14
ND0612L
(Adjunct to Oral Therapy)
Day 14
Plasma LD (ng/ml)
10000
1000
100
10
n=30
-2
3
8
Time (h)

Figure 3: ND0612L as monotherapy*

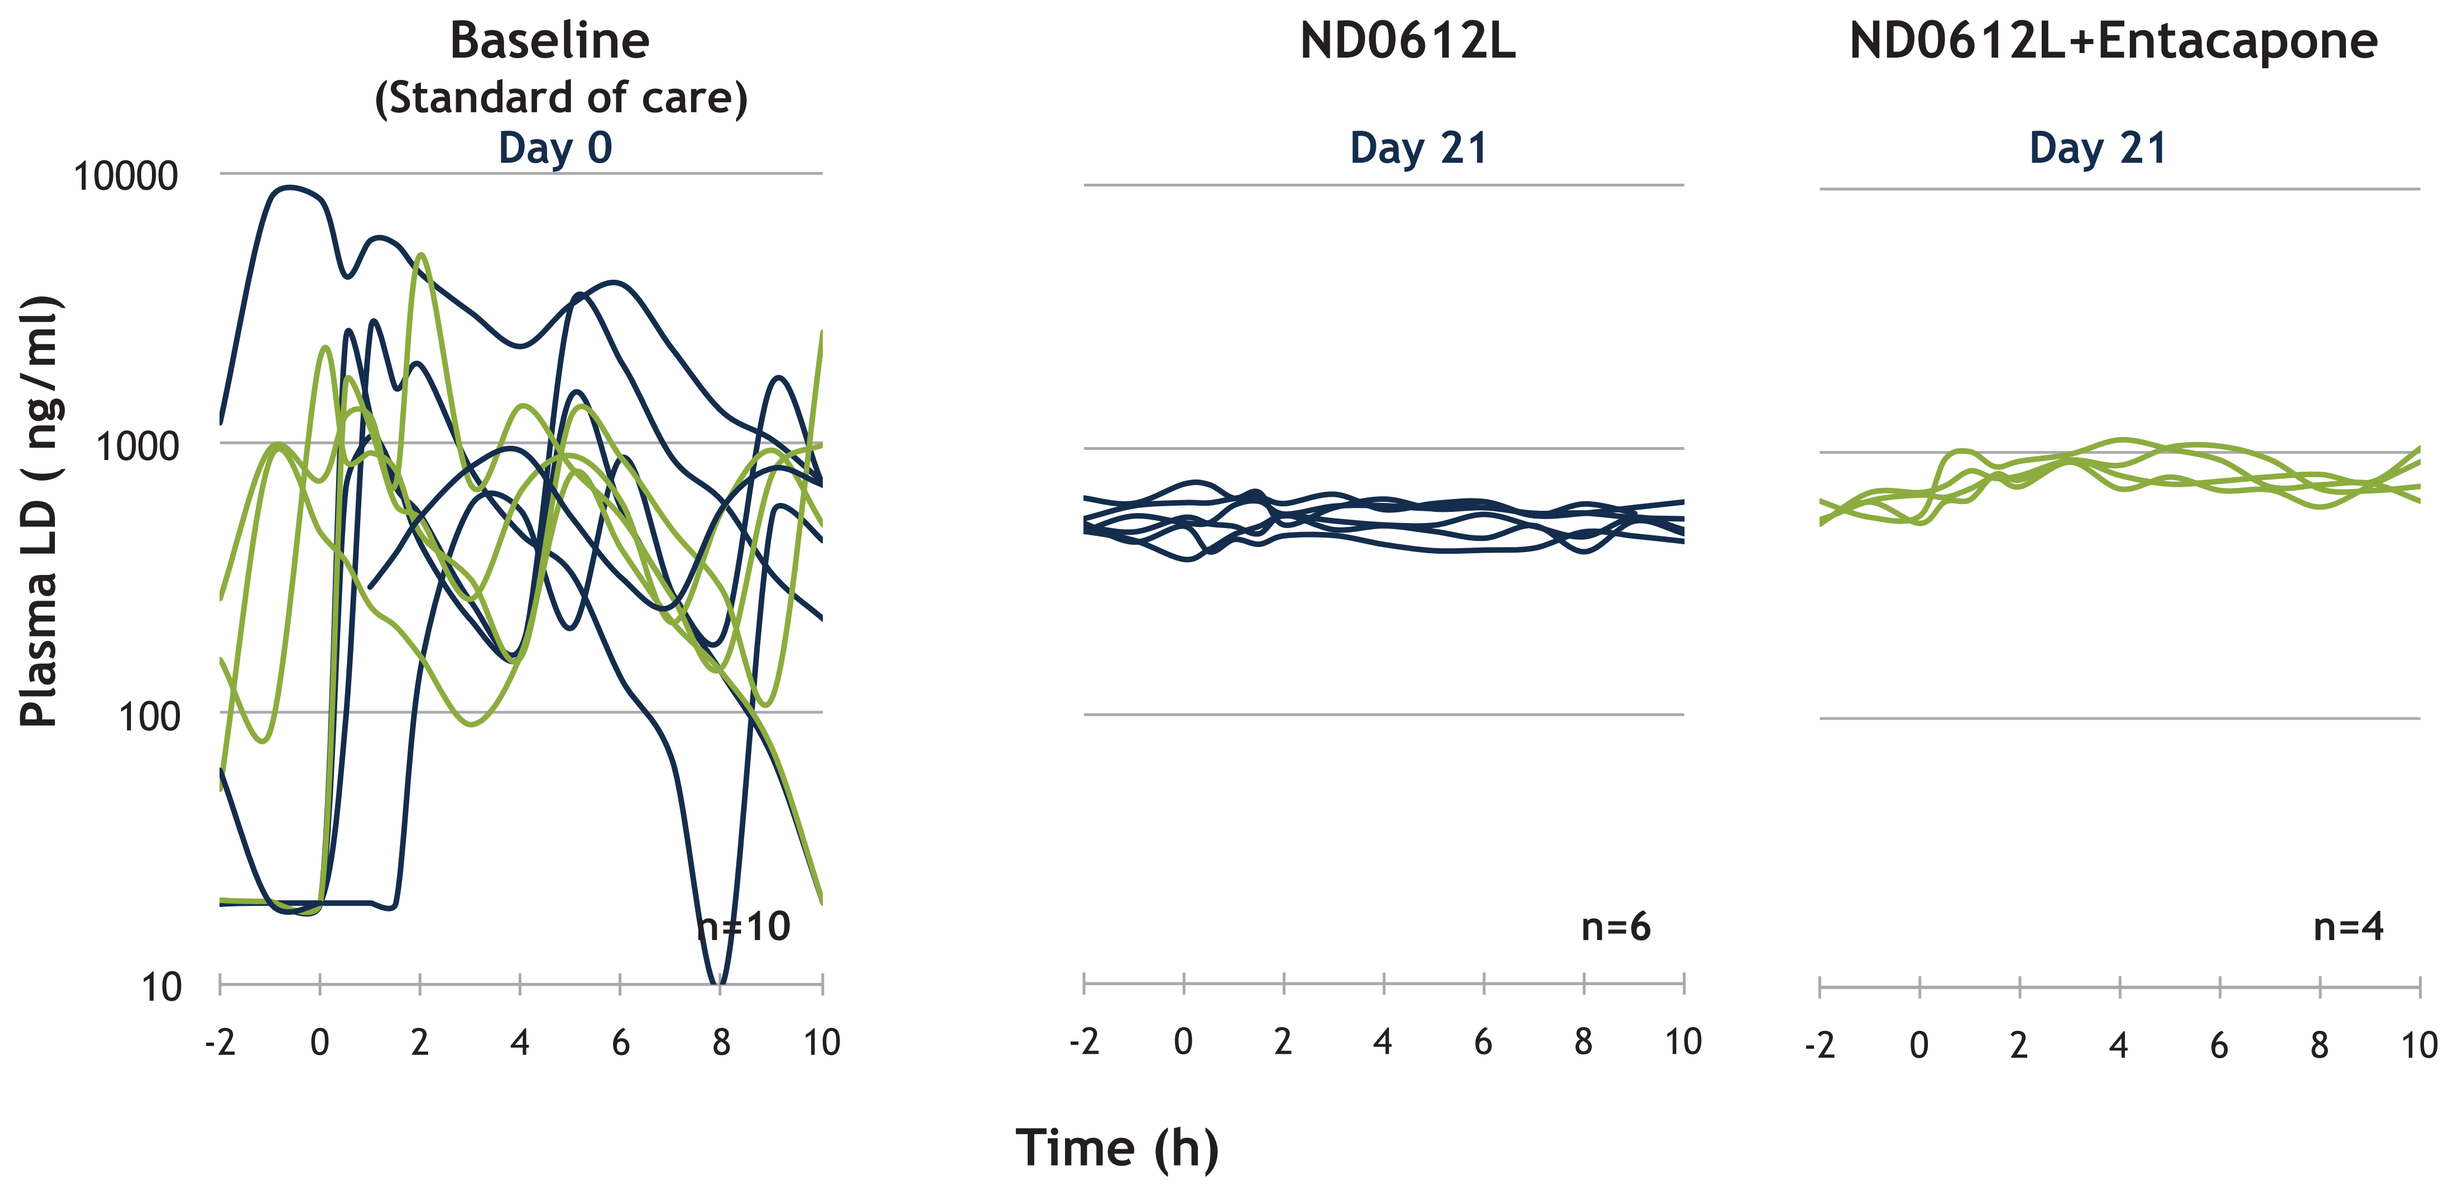
Baseline
(Standard of care)
Day 0
ND0612L
Day 21
ND0612L+Entacapone
Day 21
Plasma LD (ng/ml)
10000
1000
100
10
n=10
n=6
n=4
-2
0
2
4
6
8
10
Time (h)

Data are shown for the 9 patients who did not require additional oral LD for their PD control on the day of pharmacokinetic analysis

Exploratory efficacy

Adjunct treatment with low-dose ND0612 reduced OFF time by a mean ±SD of 2.42±2.62h and 2.13±2.24h from baseline according to in-clinic and home diaries, respectively (vs. 0.41±2.62h and 1.39±2.33h with placebo) (Figure 4).

These therapeutic effects were observed in the ND0612 treatment arm in the absence of any increases in dyskinesia, and were accompanied with improvements from baseline in other parameters including:

- Sleep quality: 17.1 ± 17.58 improvement in PDSS scores in the ND0612 group versus 0.5 ± 11.35 in the placebo group (Figure 5).
- Quality of life: 6.6 ± 10.52 improvement in PDQ-39 scores in the ND0612 group versus 1.78 ± 11.10 in the placebo group (Figure 5).
- Global clinical disease severity: 90% of patients had improved CGI-C scores in the ND0612 group versus 36% in the placebo group (Figure 5).

Figure 4: Reductions in (a) OFF time and (b) ON time spent with troublesome dyskinesia with ND0612L

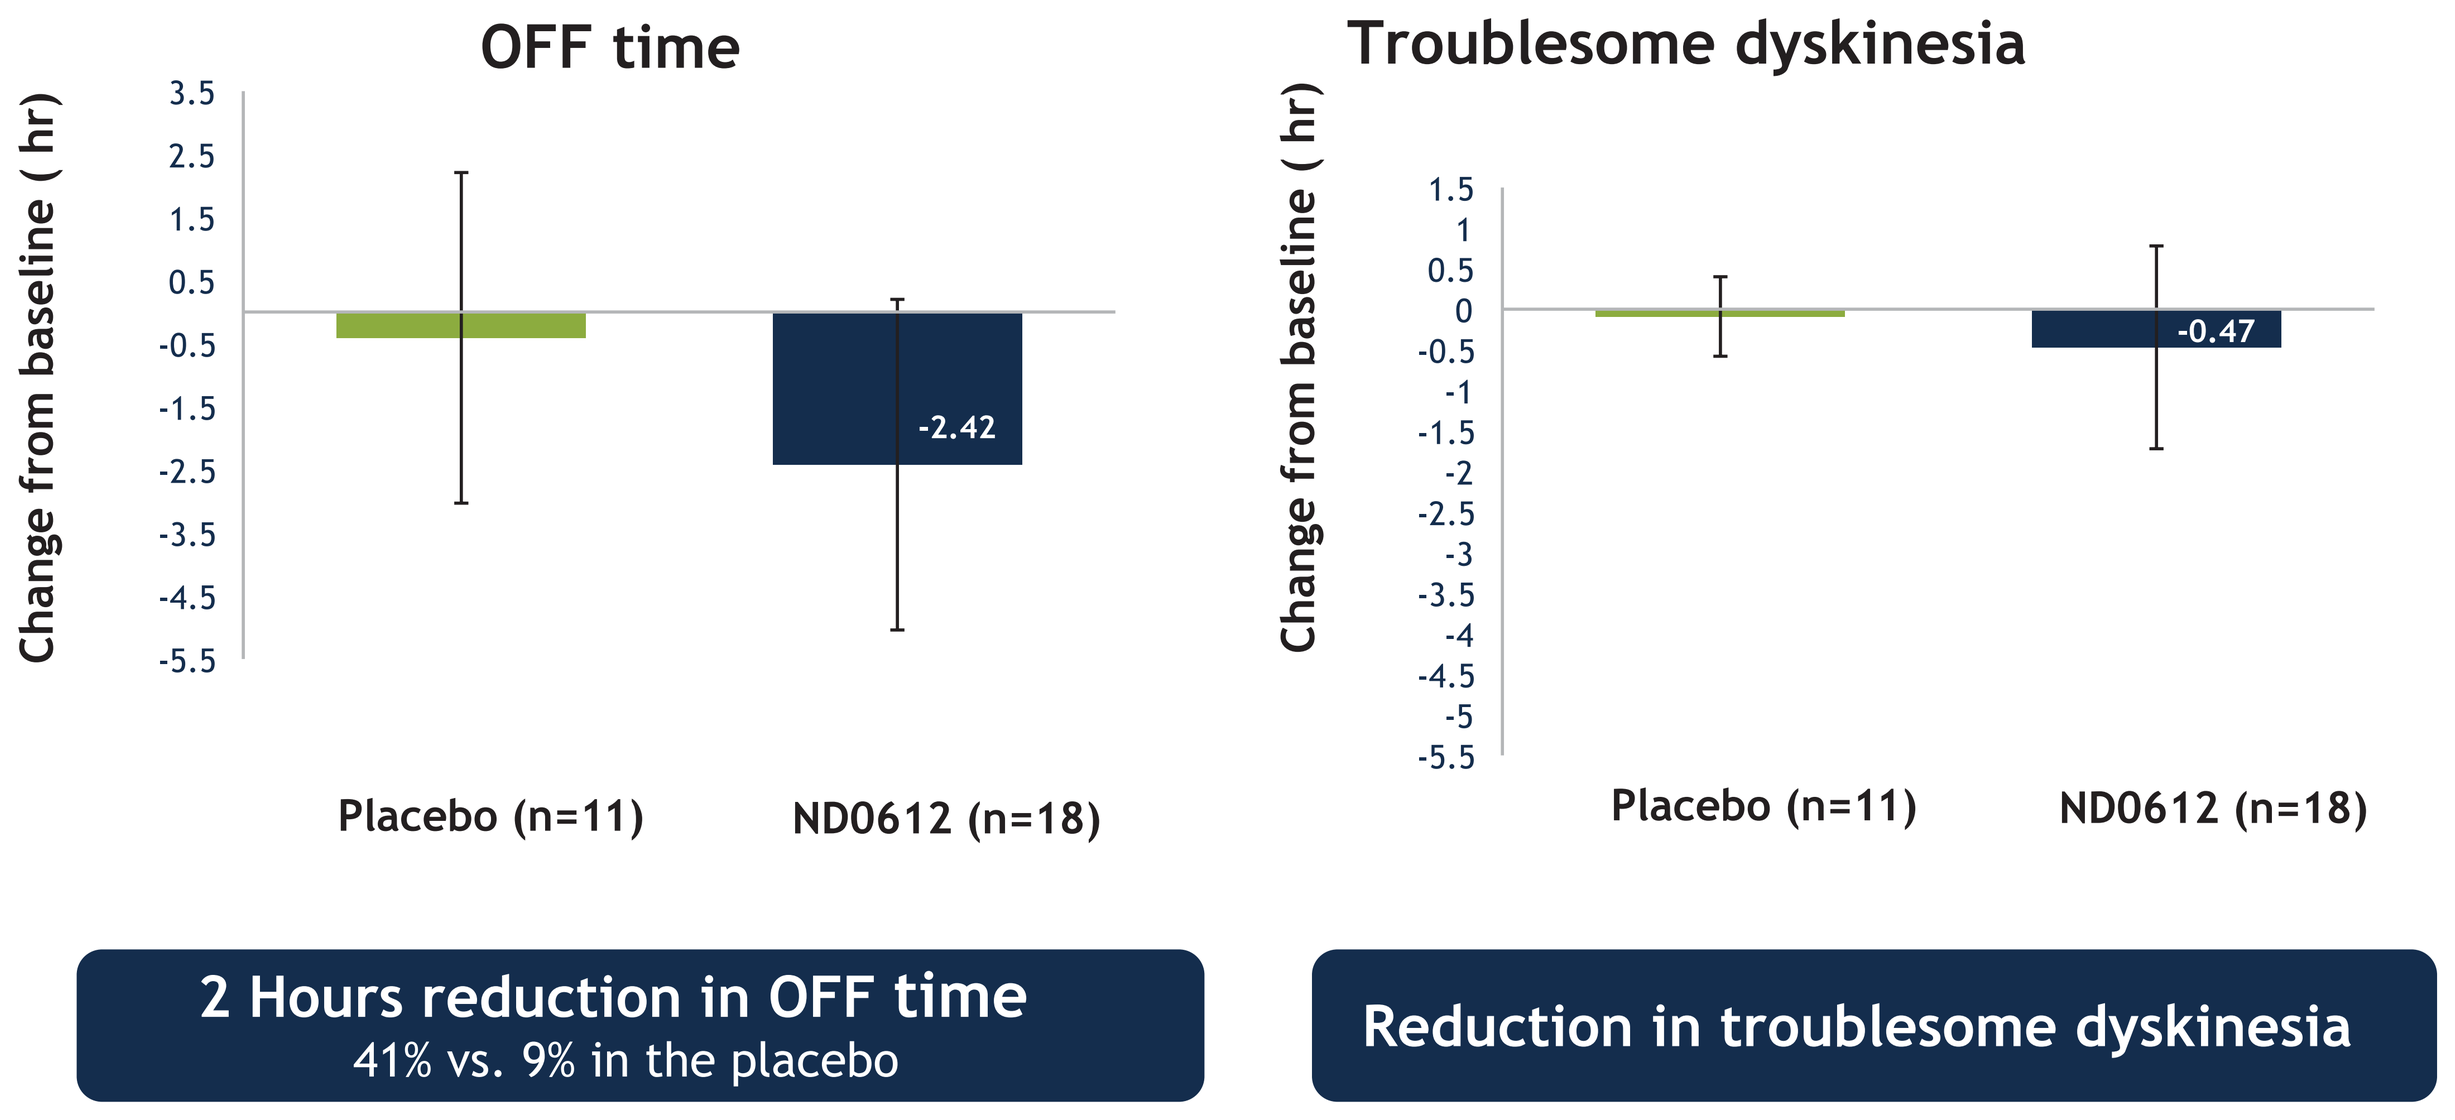
OFF time
Change from baseline (hr)
-2.42
Placebo (n=11)
ND0612 (n=18)
Troublesome dyskinesia
Change from baseline (hr)
-0.47
Placebo (n=11)
ND0612 (n=18)
2 Hours reduction in OFF time
41% vs. 9% in the placebo
Reduction in troublesome dyskinesia

Figure 5: ND0612L improves (a) quality of sleep, (b) quality of life and (c) global clinical disease severity

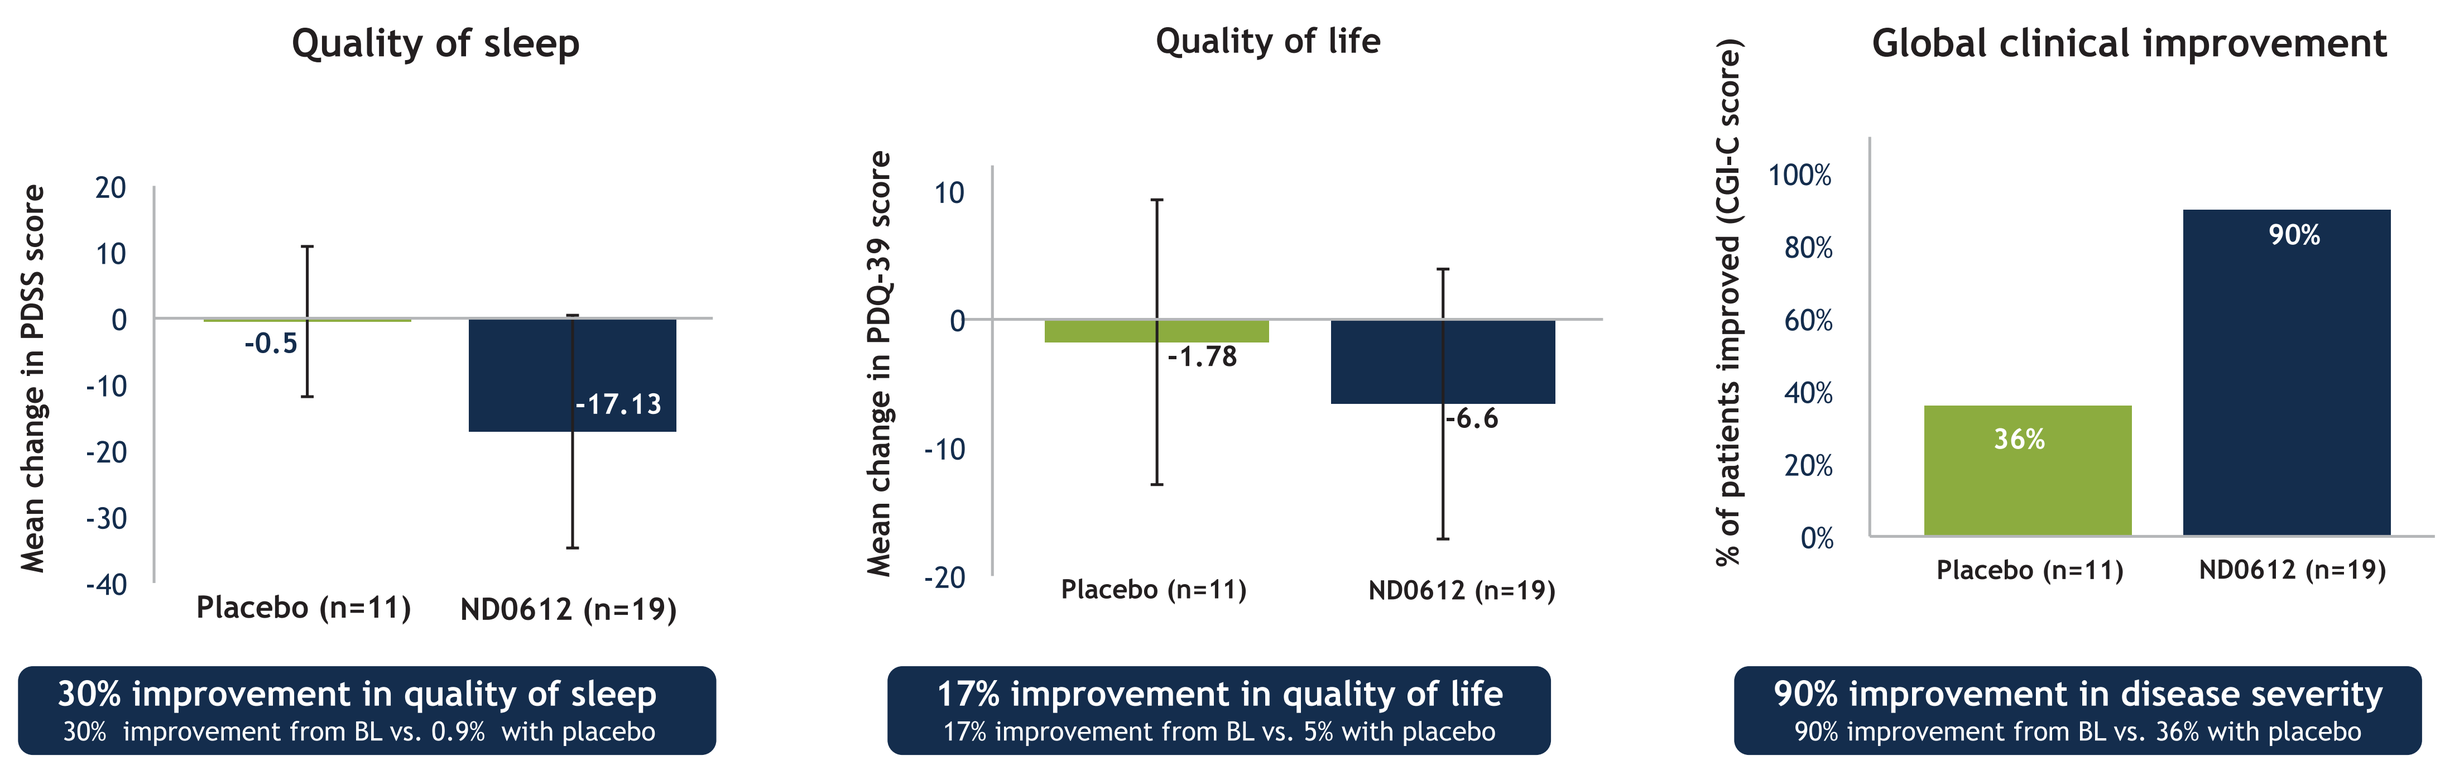
Quality of sleep
Mean change in PDSS score
-0.5
-17.13
Placebo (n=11)
ND0612 (n=19)
30% improvement in quality of sleep
30% improvement from BL vs. 0.9% with placebo
Quality of life
Mean change in PDQ-39 score
-1.78
-6.6
Placebo (n=11)
ND0612 (n=19)
17% improvement in quality of life
17% improvement from BL vs. 5% with placebo
Global clinical improvement
% of patients improved (CGI-C score)
36%
90%
Placebo (n=11)
ND0612 (n=19)
90% improvement in disease severity
90% improvement from BL vs. 36% with placebo

Safety

Study drug was well tolerated, and there were no treatment discontinuations. There were no signals suggesting that the systemic safety profile of adjunct therapy with continuous SC ND0612 infusion differs from that of oral LD/CD. Notably, there were no reports of TEAEs of dyskinesia or psychiatric symptoms. The local skin safety was characterized by occasional mild and transient infusion site reactions (erythema, edema and pain) and frequent formation of subcutaneous nodules that were mostly unfelt by patients and slowly resolved spontaneously.

Conclusions

These data suggest that subcutaneous continuous delivery of LD/CD with low-dose ND0612 provides relatively stable plasma LD levels with reduced variability compared with oral LD. Exploratory efficacy findings are also promising and warrant further study.

NeuroDerm