A randomized controlled clinical study to evaluate the efficacy and safety of ND0612, a subcutaneous infusion of levodopa/carbidopa, in patients with advanced Parkinson's disease

Karl Kieburtz[1], C. Warren Olanow[2], Yael Cohen[3] and Sheila Oren[3].

1. Clintrex, Rochester, NY, United States. 2. Clintrex, New York, NY, United States. 3. NeuroDerm Ltd., Rehovot, Israel.

Introduction

- Treatment with levodopa/dopa decarboxylase inhibitor (DDI) remains the cornerstone of Parkinson's disease (PD) management providing benefit to the majority of patients with PD. However, chronic oral treatment is associated with the development of motor complications. These complications may be related to the fluctuating levodopa (LD) blood levels, which are in part related to the short half life of LD. These motor complications with advancing disease can limit the clinical utility of LD.[1]
- Continuous levodopa/DDI delivery is considered to be the optimal method for achieving stable LD blood levels. Infusion delivery routes for treating patients with PD with advanced motor fluctuations are currently available,[1, 2] but require an invasive procedure for placement, with potentially serious complications.[3]
- Subcutaneous infusion of levodopa has the potential to provide a better tolerated and more convenient route of continuous levodopa delivery. However, poor levodopa solubility has, until now, precluded this approach.
- NeuroDerm has developed, a proprietary novel liquid formulation of levodopa/carbidopa (60 mg/mL of levodopa and 7.5 mg/mL of carbidopa) delivered by subcutaneous infusion by a belt pump system, which has been specifically developed to provide a continuous and stable level of levodopa in patients with moderate and advanced PD.
- ND0612 is developed in 2 dosages: a low dose - of up to 360 mg levodopa that is infused subcutaneously via one infusion site, and a high dose - ND0612H of up to 720 mg levodopa that is infused subcutaneously via two infusion sites.

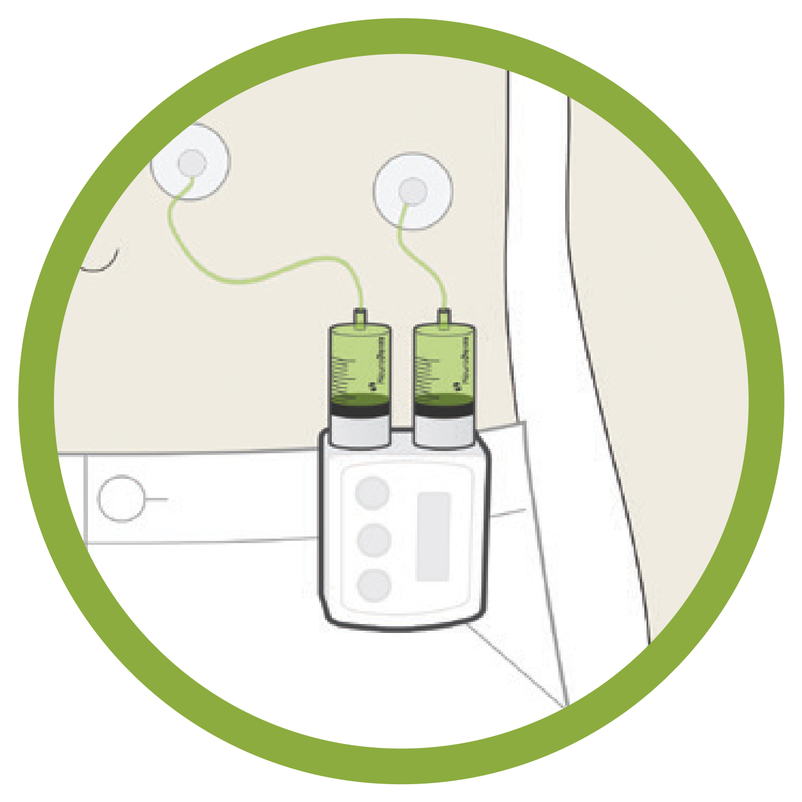

ND0612 Twin pump

- Previous studies have demonstrated that continuous subcutaneous delivery of high dose ND0612 has the capacity to produce stable plasma levodopa levels, comparable to those attained by treatments that require invasive surgery.[4] In these studies, treatment with ND0612 was well tolerated. Infusion site reactions were mild and transient.
- In addition, continuous 'round the clock' dopaminergic delivery (24 hours) may offer a benefit over delivery only during the waking day. It has been previously suggested that 24 hour delivery may help avoid morning akinesia,[5] which can contribute to poorer quality of life in patients with advanced PD.[6]

Objective

- The primary objective of this study is to assess the effect of continuous subcutaneous infusion of two dosing regimens (24-hours and 14-hours infusion with a morning oral dose of LD/CD) of high dose ND0612 on daily OFF time.
- To assess the difference between the 2 continuous subcutaneous infusion dosing regimens of high dose ND0612 vs. standard oral LD/DDI treatment on ability to induce early morning full ON.

Methods

Study design

- This is a multi center, parallel-group, rater-blinded, randomized, Phase II clinical study (NCT02577523) in patients with advanced PD investigating the efficacy, pharmacokinetics, safety and tolerability of continuous subcutaneous infusion of two dosing regimens of high dose ND0612 compared to standard oral levodopa/DDI.
- Patients will start the inpatient phase after discontinuing oral LD/DDI the night before. All patients will undergo 1 day of standard oral levodopa/DDI inpatient dosing followed by 2 days of inpatient treatment with either a 24-hours or 14-hours ND0612 infusion (1:1 randomization ratio) regimen.
- Patients will know which treatment regimen they are assigned to, as will the unblinded staff responsible for administering the study treatments. However, all assessments of motor state and time to full ON will be performed by a blinded rater.
- Patients will then continue as outpatients on a maintenance dose of the assigned ND0612 dosing regimen for the next 25 days.
- An additional safety visit will be performed 4 weeks after the last administration of ND0612.

Patients

- Planned recruitment of 36 subjects (18 per arm) from 10 study sites.
- Male and female (30 – 80 years) patients with a diagnosis of PD according to UK Brain Bank Criteria.[7]
- Hoehn & Yahr (during ON) of stage ≤3, with ≥2.5 hours of OFF time per day.
- Predictable and well defined early morning OFF periods and a 'good' early morning response to levodopa.

Study design

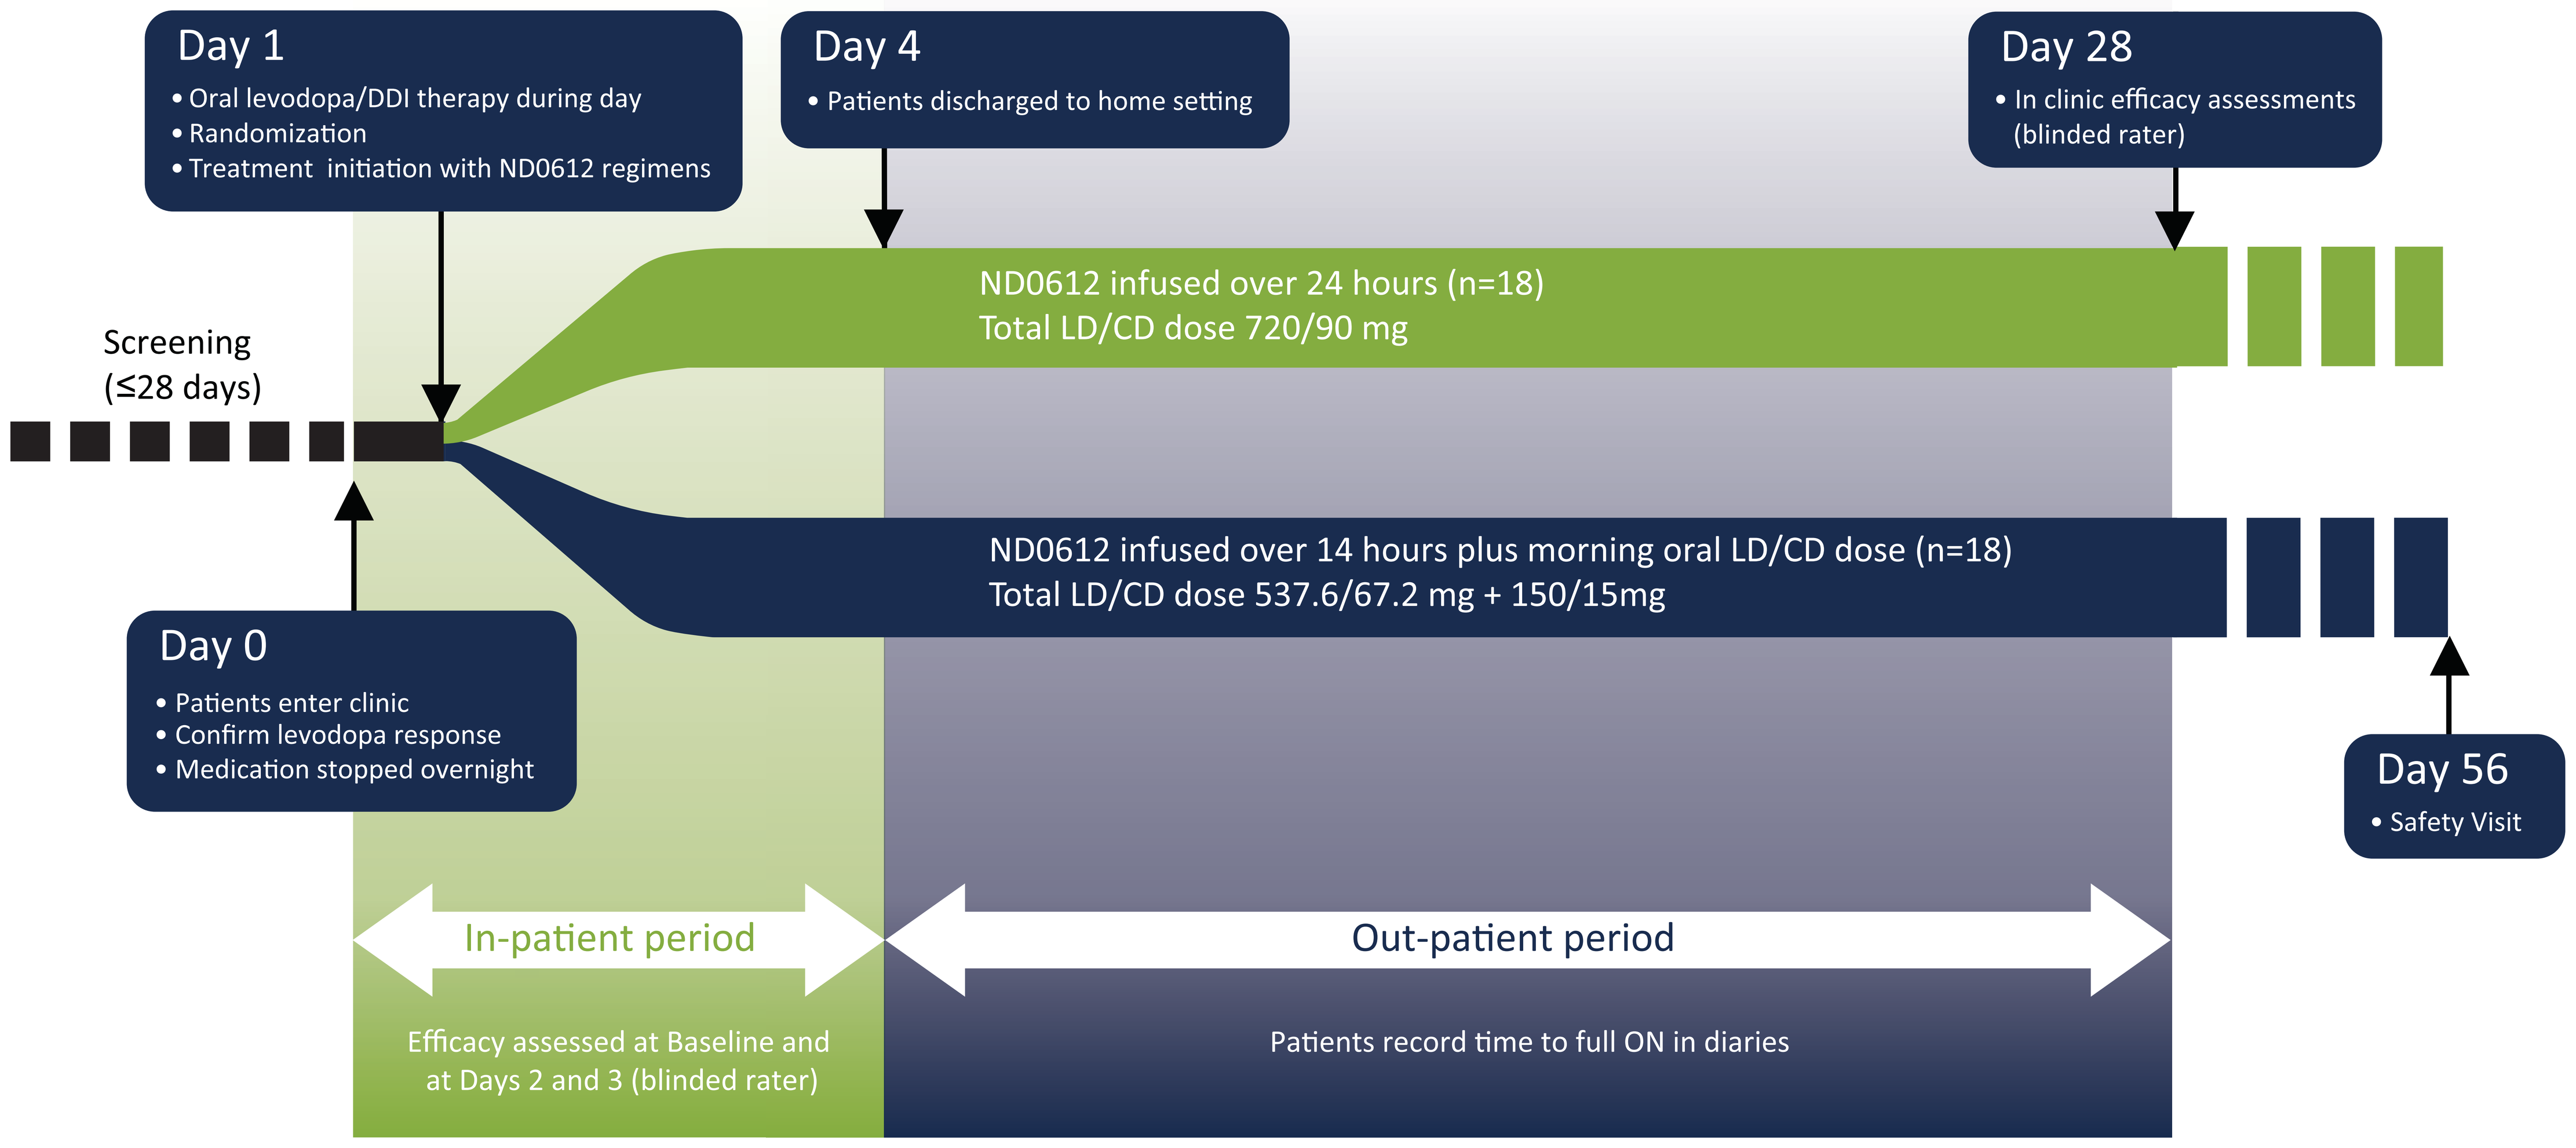
Day 1
• Oral levodopa/DDI therapy during day
• Randomization
• Treatment initiation with ND0612 regimens
Day 4
• Patients discharged to home setting
Day 28
• In clinic efficacy assessments (blinded rater)
Screening (≤28 days)
ND0612 infused over 24 hours (n=18)
Total LD/CD dose 720/90 mg
ND0612 infused over 14 hours plus morning oral LD/CD dose (n=18)
Total LD/CD dose 537.6/67.2 mg + 150/15mg
Day 0
• Patients enter clinic
• Confirm levodopa response
• Medication stopped overnight
Day 56
• Safety Visit
In-patient period
Out-patient period
Efficacy assessed at Baseline and at Days 2 and 3 (blinded rater)
Patients record time to full ON in diaries

- Patients should be taking ≥4 levodopa doses per day (≥3 doses/day of Rytary) and have attempted to take at least 2 other classes of PD medications in a therapeutic dose for at least 30 consecutive days each. All PD medications should be stable for ≥30 days prior to the start of the study.
- Prior exposure to subcutaneous apomorphine injections/infusion is permitted but patients must have stopped administration ≥4 weeks before the screening visit. Treatment with apomorphine is prohibited during the entire ND0612 treatment period.
- Key exclusion criteria include: atypical parkinsonism, a Mini Mental State Examination (MMSE) score ≤26 and any relevant medical, surgical, or psychiatric condition (including acute psychosis or hallucinations in prior 6 months), laboratory value, or concomitant medication which could compromise safety or potentially prevent completion of all aspects of the study. Patients who have undergone prior neurosurgical procedure for PD, or Duodopa treatment are not eligible for inclusion in this study.
- All patients should have a named study partner (e.g. spouse, relative, friend, neighbor, or caregiver) who is willing and able to be support the subject during the study period.

Study Treatment

- This will be the first study to assess the administration of high dose ND0612 in the home setting. A Home Nursing Service representative will visit the subjects at their homes and will supervise the administration of the infusion and operation of the pump system.
- Patients randomized to the 24-hours ND0612 group will receive a total levodopa/carbidopa dose of 720/90 mg. Rates of infusion will be higher in the daytime (18 hours; 0.64 mL/hr) and lower in the night (6 hours; 0.08 mL/hr). Subjects will wear the pump during sleep.
- Patients randomized to the 14-hours ND0612 group will receive a total levodopa/carbidopa dose of 537.6/ 67.2mg during the waking hours, and will disconnect the pump during sleep. Patients in this group will take a supplemental dose of oral levodopa/carbidopa (150/15 mg) in the morning.
- Study treatment is administered by an infusion pump which is connected to 2 cannulas that are inserted to the subcutaneous layer of the skin.
- The infusion sites are changed every morning in a rotating manner.
- The recommend infusion sites are the abdomen (including flanks), and outer thighs.

Assessments

Efficacy • OFF time • Percentage of subjects with full ON at 8 am and 9 am • ON with and without dyskinesia • UPDRS (total, motor and activities of daily living) scores • Clinical Global Impression (Improvement and Severity) • Parkinson's Disease Sleep Scale • Need for dose adjustments of oral PD medication	**Safety/Tolerability** • AE reporting • Local assessment of infusion sites (e.g. nodule formation, pain) • Daytime sleepiness (Epworth Sleepiness Scale) • Impulse control tolerability (Questionnaire for Impulsive-Compulsive Disorders in Parkinson's Disease-Rating Scale) • Suicidality (Columbia Suicide Severity Rating Scale) • Laboratory parameters, vital signs, physical and neurological examination • Electrocardiogram (ECG) evaluation
Patient reported outcomes • Subject's Global Impression of Improvement • Satisfaction with the dosing regimen • Night-time sleep • Need for dose adjustments of oral PD medication	**PK** • Venous sampling for pharmacokinetic assessments will be performed on Days 2, 3 and 28 (at 2 and 4 hours post dose)

Statistical analyses

- The primary efficacy analysis will include all randomized subjects who have scoring of motor activity at baseline and on Day 3 or Day 28. Safety analyses will include all subjects who received ≥1 dose of ND0612.
- The primary endpoint is the change in OFF time from Baseline to Day 28 within within each treatment regimen and the difference between the 2 treatment regimens estimated using a Mixed Model for Repeated Measures (MMRM) including response data from Day 3 and 28 with no imputation for missing data.
- The key secondary analysis will compare the percentage of subjects in each group achieving full ON (defined as ON response comparable to that achieved with oral levodopa/DDI treatment) in the morning.

Summary

- Patients with advanced PD who suffer motor complications that cannot be adequately controlled with current therapy face limited treatment options that include surgical intervention that may be associated with serious adverse effects.
- This will be the first efficacy trial of high dose ND0612, marking an important milestone in its clinical development.
- In the present study we compare continuous infusion round the clock with infusion only during the waking day with a morning oral bolus of LD/CD.
- Patients who complete the treatment period of the current study will have the option to directly roll-over to an open-label, long-term, safety study.

References

1. Olanow CW, Stern MB, Sethi K. The scientific and clinical basis for the treatment of Parkinson disease (2009). Neurology 2009;72(21 Suppl 4):S1-136.
2. LeWitt PA. Levodopa therapy for Parkinson's disease: Pharmacokinetics and pharmacodynamics. Mov Disord 2015;30(1):64-72.
3. Olanow CW, Kieburtz K, Odin P, et al. Continuous intrajejunal infusion of levodopa-carbidopa intestinal gel for patients with advanced Parkinson's disease: a randomised, controlled, double-blind, double-dummy study. Lancet Neurol 2014;13(2):141-149.
4. Giladi N, Caraco Y, Gurevich T, et al. Pharmacokinetic profile of ND0612 (levodopa/carbidopa for subcutaneous infusion) in Parkinson's disease (PD) patients with motor fluctuations: results of a Phase IIa dose finding study (Abstract). Mov Disord 2015;30 (Suppl 1).
5. Trenkwalder C, Kies B, Rudzinska M, et al. Rotigotine effects on early morning motor function and sleep in Parkinson's disease: a double-blind, randomized, placebo controlled study (RECOVER). Mov Disord 2011;26(1):90-99.
6. Chapuis S, Ouchchane L, Metz O, Gerbaud L, Durif F. Impact of the motor complications of Parkinson's disease on the quality of life. Mov Disord 2005;20(2):224-230.
7. Hughes AJ, Daniel SE, Kilford L, Lees AJ. Accuracy of clinical diagnosis of idiopathic Parkinson's disease: a clinico-pathological study of 100 cases. J Neurol Neurosurg Psychiatry 1992;55(3):181-184.

NeuroDerm

Disclosures: Nir Giladi, Yoseph Caraco, Tanya Gurevitch and Ruth Djaldetti report personal compensation for speaking and/or consultative services from Neuroderm.Yael Cohen, Oron Yacobi-Zeevi and Sheila Oren are employed by Neuroderm